Novel Polyvalent Ionizable Lipids Enable Targeted Delivery of mRNA to T Cells and Monocytes

FACTOR® BIOSCIENCE

Ariadna Lubinus[1], Joseph Pisano[1], Christopher B. Rohde[1], Matthew Angel[1]

[1]Factor Bioscience Inc., Cambridge MA

This work is protected by one or more pending patent applications.

Introduction

Ionizable lipids are established nucleic acid delivery vehicles capable of delivering mRNA into the cytosol of target cells to induce protein expression. Although risks of mutagenesis limit the clinical use of viral vectors and DNA delivery systems, mRNA delivery systems do not carry these risks. However, lipid-based mRNA delivery systems often suffer from low transfection efficiency, especially for cell types such as primary cells and immune cells which are adept at recognizing and rejecting foreign nucleic acids.

To address this limitation, we designed a library of 4,200 ionizable lipids of which a panel of 84 candidates were synthesized and screened in human peripheral mononuclear cells (PBMCs) and other immune cells including THP-1 monocytes. Human PBMCs were transfected with GFP-mRNA formulated into complexes with the candidate lipids. 43 of these lipids possess a spermine-derived backbone, a structural characteristic of the ToRNAdo™ Nucleic-Acid Delivery System which has delivered mRNA to multiple cell types ex vivo and in vivo.

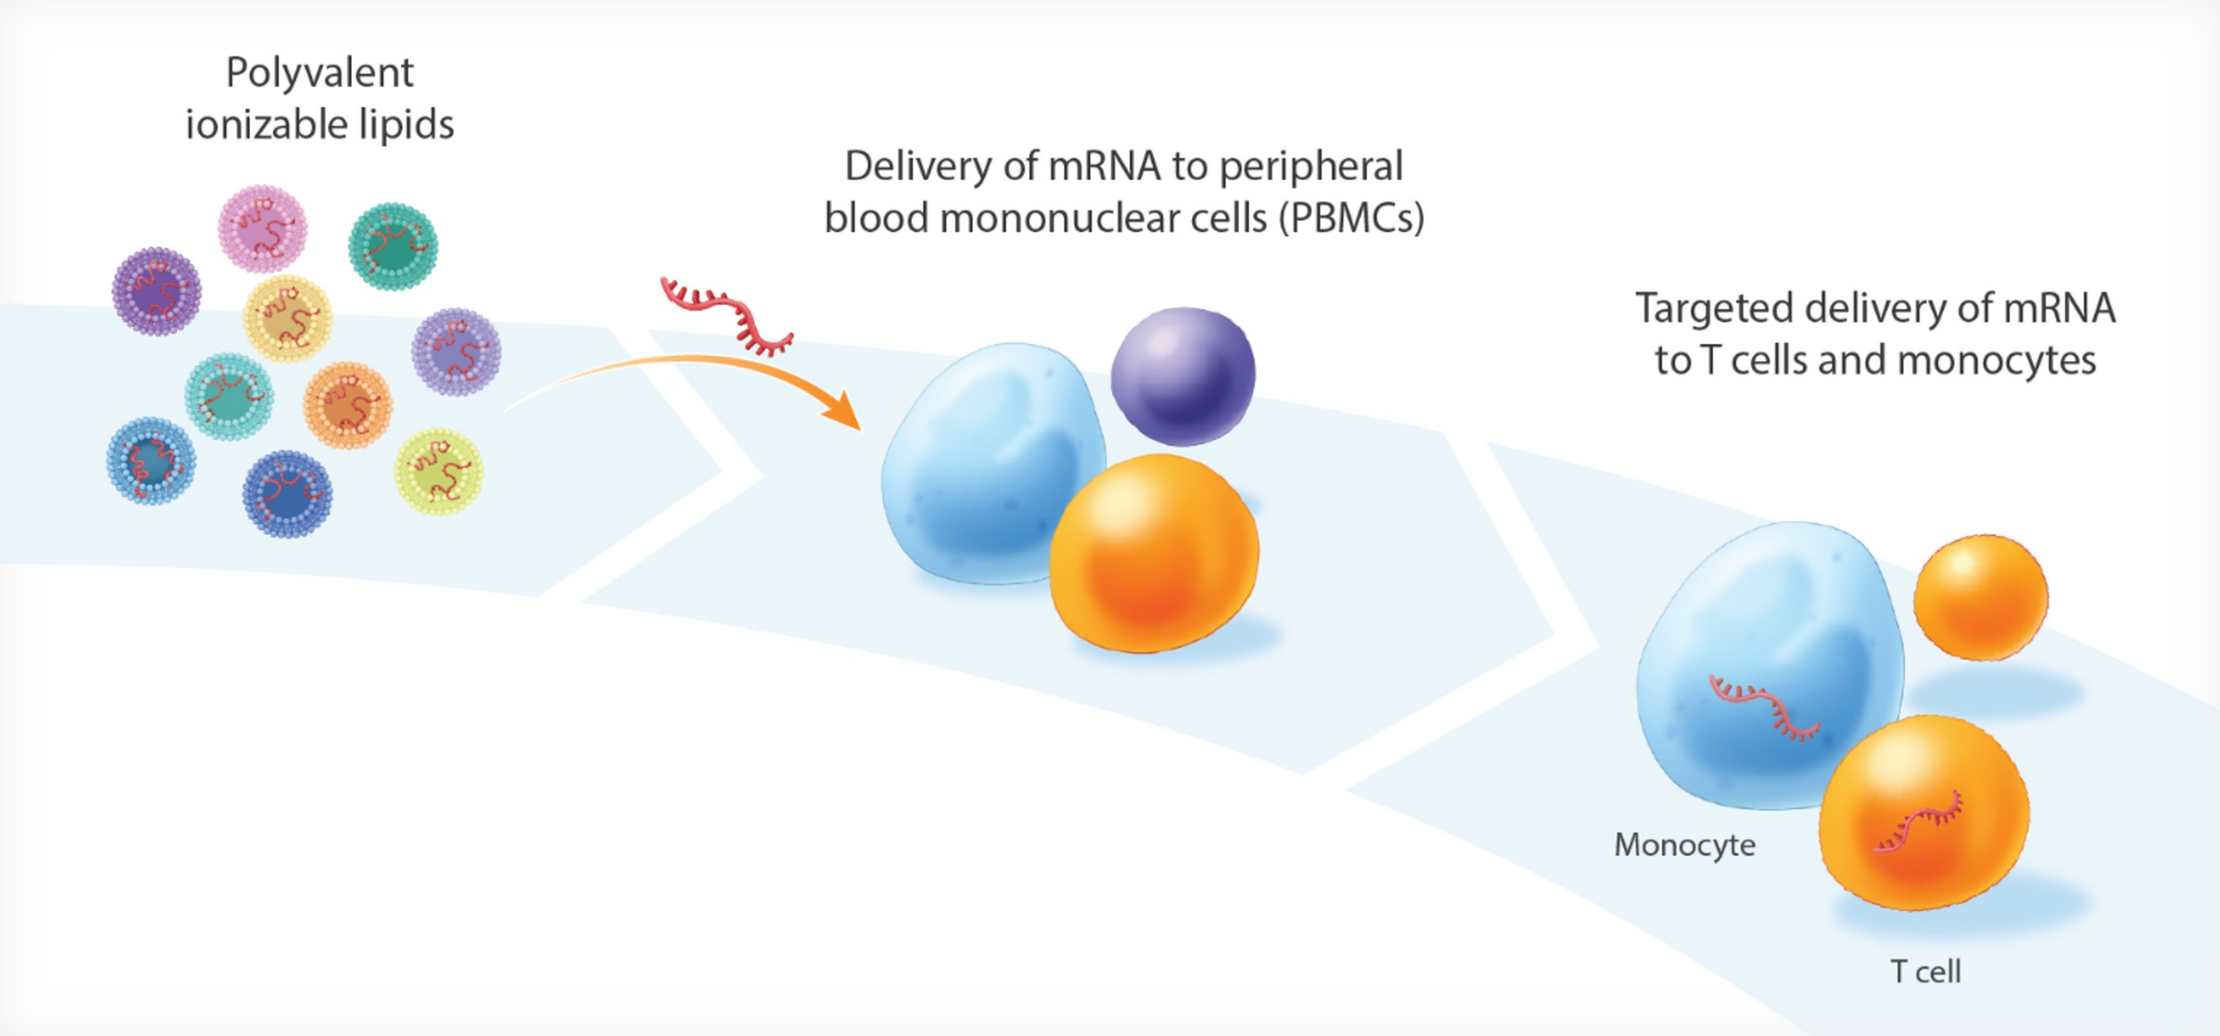
Polyvalent ionizable lipids
Delivery of mRNA to peripheral blood mononuclear cells (PBMCs)
Targeted delivery of mRNA to T cells and monocytes
Monocyte
T cell

Results

PBMCs were treated with 1.33 ng/uL of GFP mRNA per 100,000 cells at a lipid:mRNA ratio (w/w) of 3:1. Cellular imaging showed peak GFP fluorescence intensity 18-24 hrs after transfection of lipoplexes and 24-36 hrs after LNP transfection. Of the lipids tested, those containing spermine headgroups and hexyldecanoate ester tails yielded high levels of GFP expression in CD3+ cells with a four- to eleven-fold increase in GFP positive cells relative to Lipofectamine™ 3000. The same lipids transfected THP-1 monocytes at over 90% efficiency.

Conclusion

Our data indicate that spermine-derived polyvalent ionizable lipids are able to deliver mRNA to primary human immune cells, particularly T cells and monocytes. These novel lipids may prove useful in developing mRNA therapeutics to modulate protein expression in immune cells and mediate cellular host-defense response.

1 Lipid Library Design

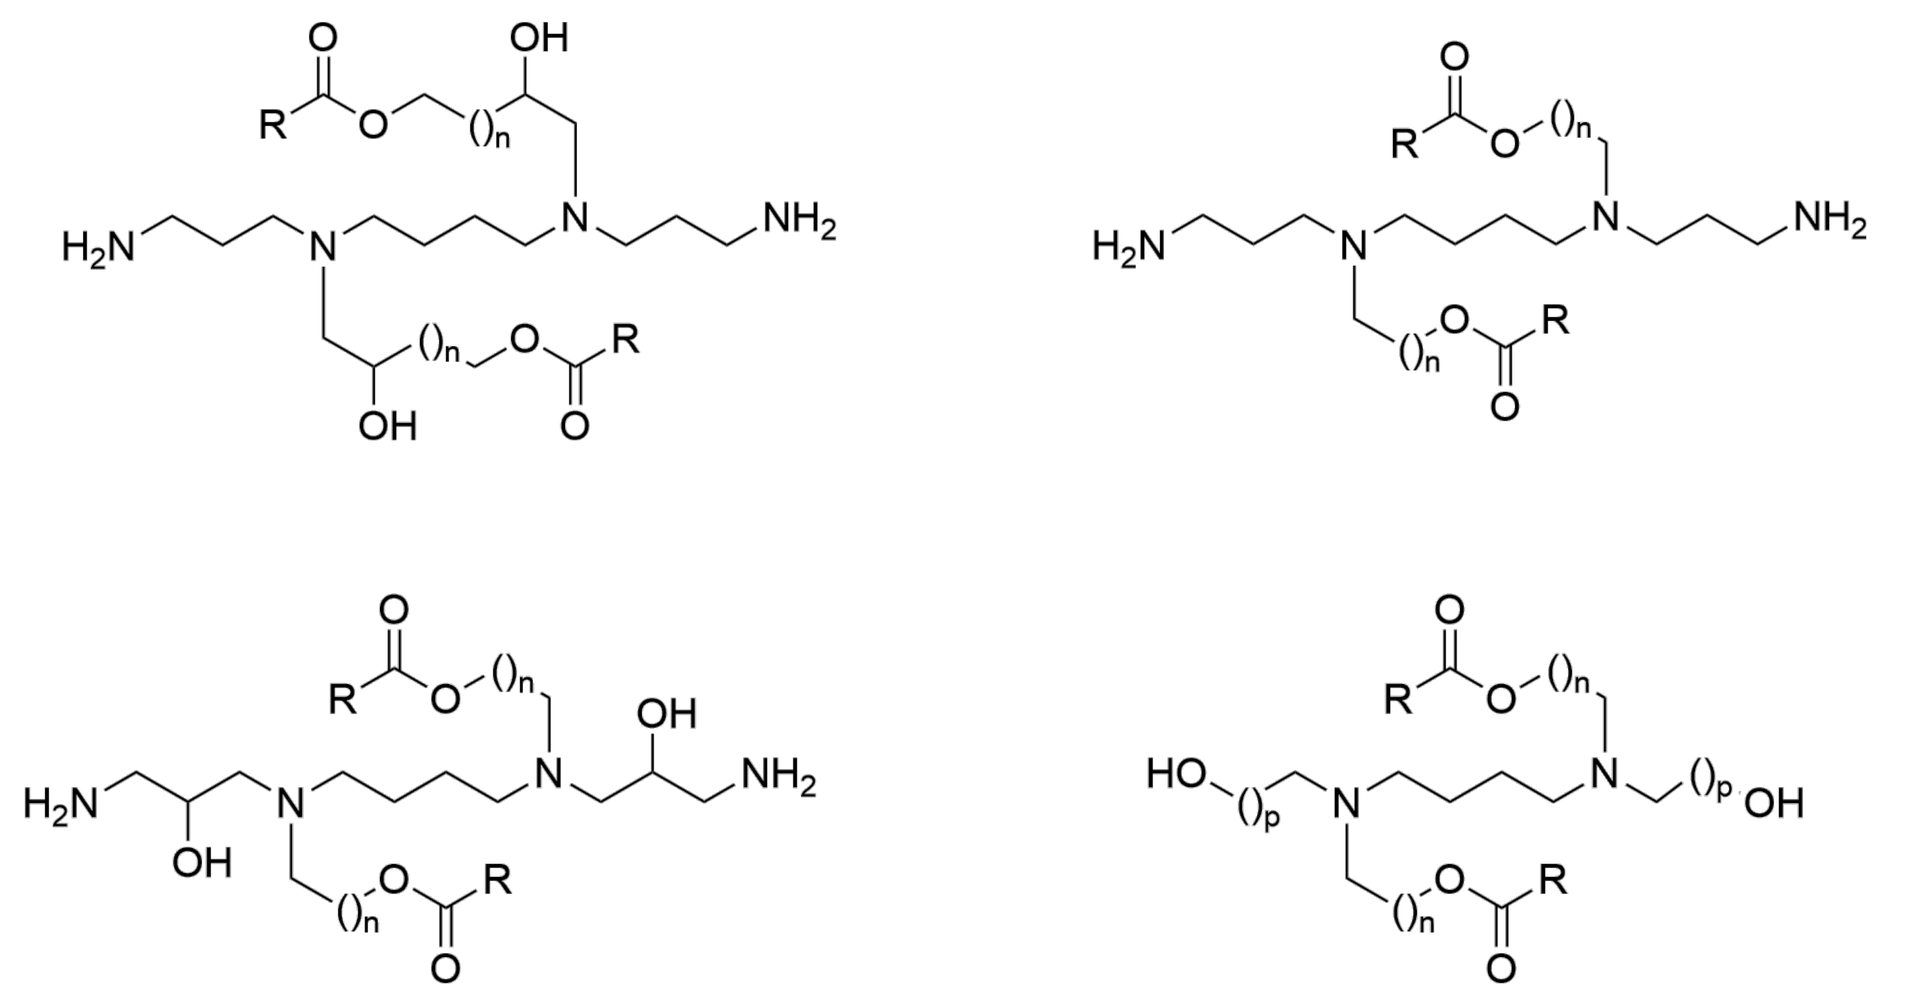

Figure 1. PBMC lipid library design. The novel ionizable lipids that make up the library contain 4 major structural variations with polyvalent headgroup designs inspired by the ToRNAdo™ lipid delivery system. Esters and hydroxyls were incorporated to promote in vivo biodegradability and increase lipid-mRNA formulation stability. "R" in the structures denotes alkyl or alkenyl branched lipid tails containing 10 to 24 carbon atoms. Tethered ester chains range from n=2 to n=6 and tethered primary alcohols range from p=1 to p=3.

2 Lipid-mRNA Delivery System

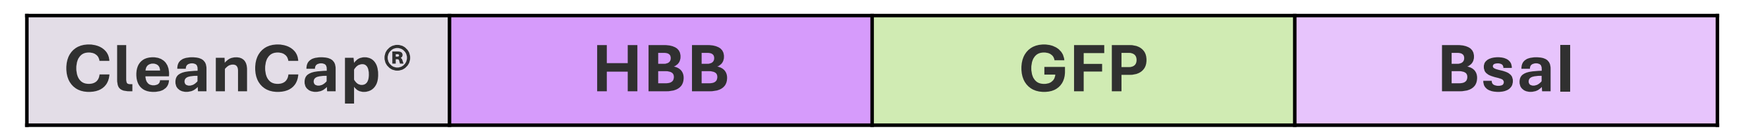
CleanCap®
HBB
GFP
BsaI

Figure 2. Structure of mRNA cargo - CleanCap® EGFP 5Hbb_CC, 3Hbb TT-BsaI. The mRNA cargo of the lipid delivery system consists of a 1093 base long sequence with an HBB promoter and GFP reporter. The mRNA is 353,070 g/mol and contains 1093 phosphates per mole.

Lipoplex Preparation

Materials OptiMEM buffer (pH 7.4), GFP mRNA, and the ionizable lipid transfection reagent (in EtOH). A 3:1 weight ratio of ionizable lipid to RNA was used for most transfections.

Complexation Lipid and RNA were mixed separately in buffer. Reagent was pipette-mixed into RNA tube, lipid-RNA solution was incubated for 5 minutes before adding dropwise into cell culture.

Lipid-RNA Nanoparticle Preparation

LNP Materials mRNA in 100 mM citrate buffer (pH 3.0), [ionizable lipid, C, DSPC, and PEG_{2000}-DMG] at [50 38.5 : 10 : 1.5 M] in EtoH

Microfluidic Formulation with NanoAssemblr® Benchtop

- 3:1 volume ratio of aqueous:organic phase
- Total lipid/mRNA weight ratio of approximately 40
- Flow rate of 3 mL/min
- Post-formulation RNA concentration of 0.2mg/mL
- Total lipid concentration of 2mg/mL and N/P of 6

Dialysis and Purification Materials 20 mM Tris buffer (pH 7.5), Slide-A-Lyzer G3 Dialysis Cassette, Amicon Ultra filters (10 kDa MWCO), 0.22 µm syringe filter membrane

Cryopreservation Conditions Buffer: 20 mM Tris + 10% sucrose, Temp: 4°C for up to 1 week; -80°C after snap-freezing in LN2

Characterization DLS, Ribogreen assay

Transfection LNPs were diluted in culture medium (with serum)

3 PBMC Lipid Library Screen

PBMC Cell Subsets	
CD3 (35-75%)	T-Cells
CD14 (10-20%)	Monocytes/Macrophages
CD19 (~15%)	B Cells
CD34 (0.1-0.2%)	HSCs and progenitor cells
CD56 (~15%)	NK Cells

Table 1. Average expression of markers in a PBMC population.

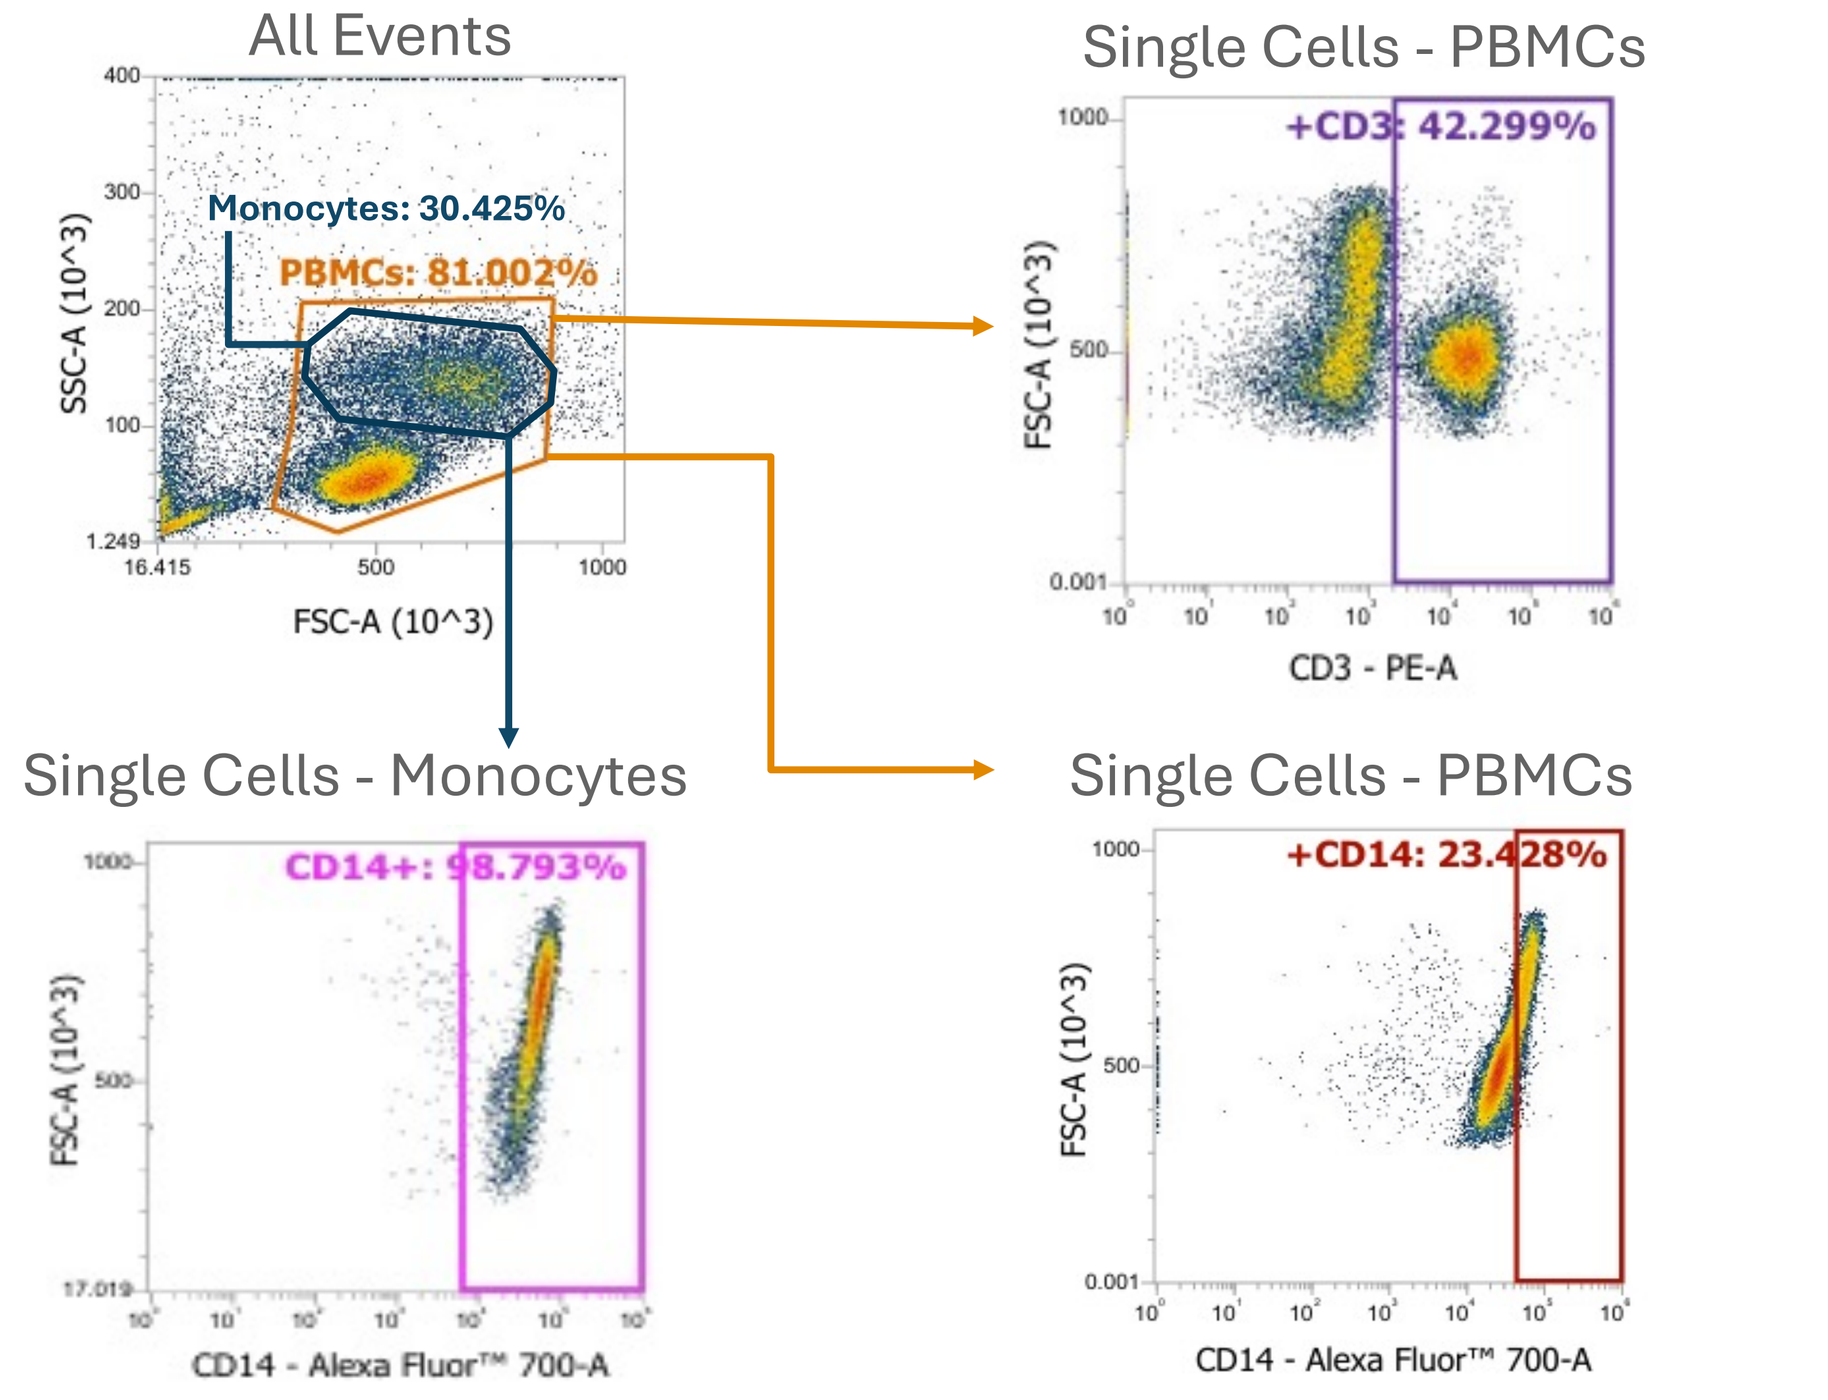
All Events
Monocytes: 30.425%
PBMCs: 81.002%
SSC-A (10^3)
FSC-A (10^3)
Single Cells - PBMCs
+CD3: 42.299%
CD3 - PE-A
Single Cells - Monocytes
CD14+: 98.793%
Single Cells - PBMCs
+CD14: 23.428%
CD14 - Alexa Fluor™ 700-A

Figure 3. PBMC flow cytometric phenotyping. Expression of CD3 (a T cell marker) and CD14 (a monocyte cell marker) were evaluated using flow cytometry to validate the panel.

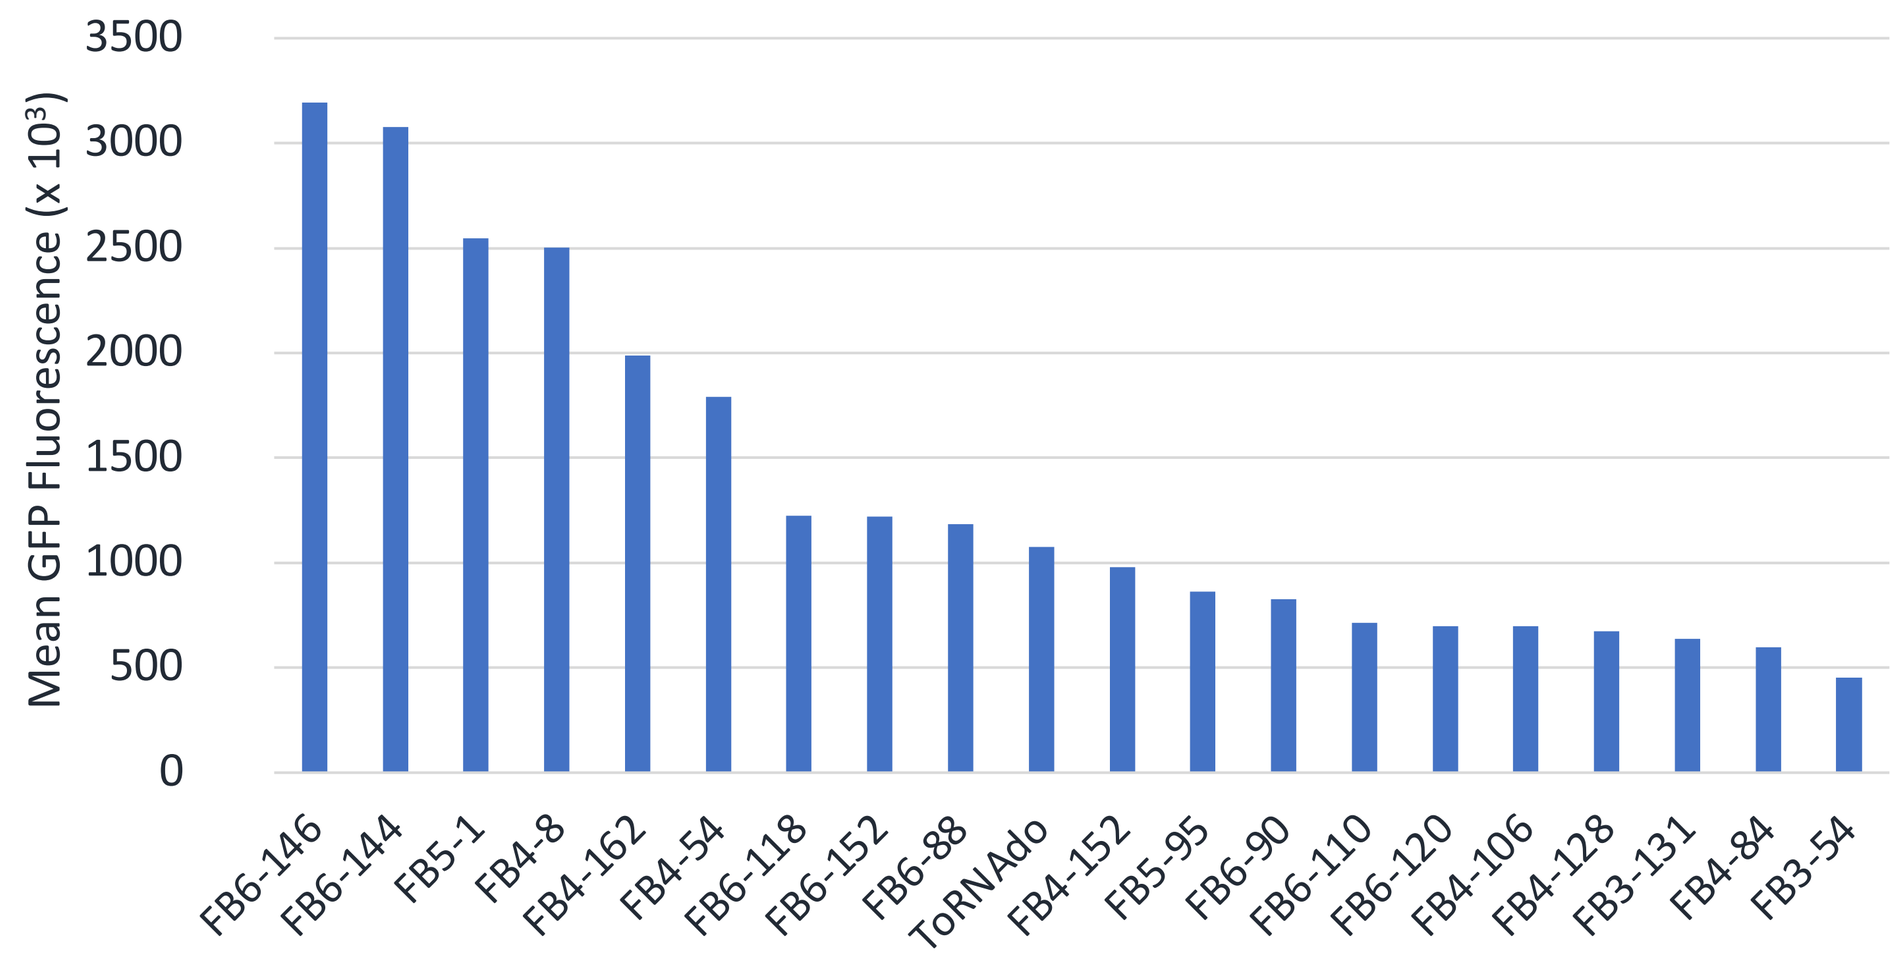
Mean GFP Fluorescence (x 10³)
FB6-146
FB6-144
FB5-1
FB4-8
FB4-162
FB4-54
FB6-118
FB6-152
FB6-88
ToRNAdo
FB4-152
FB5-95
FB6-90
FB6-110
FB6-120
FB4-106
FB4-128
FB3-131
FB4-84
FB3-54

Figure 4. Mean GFP fluorescence in PBMC lipoplex library screen. The full lipid library was prepared as lipoplexes and delivered 0.2ug GFP-RNA to 100,000 PBMCs per sample. The 20 lipids exhibiting the highest mean GFP fluorescence, calculated as number of GFP+ cells in the sample multiplied by sample mean fluorescent intensity, is plotted for the peak expression time of 18 hrs post-transfection.

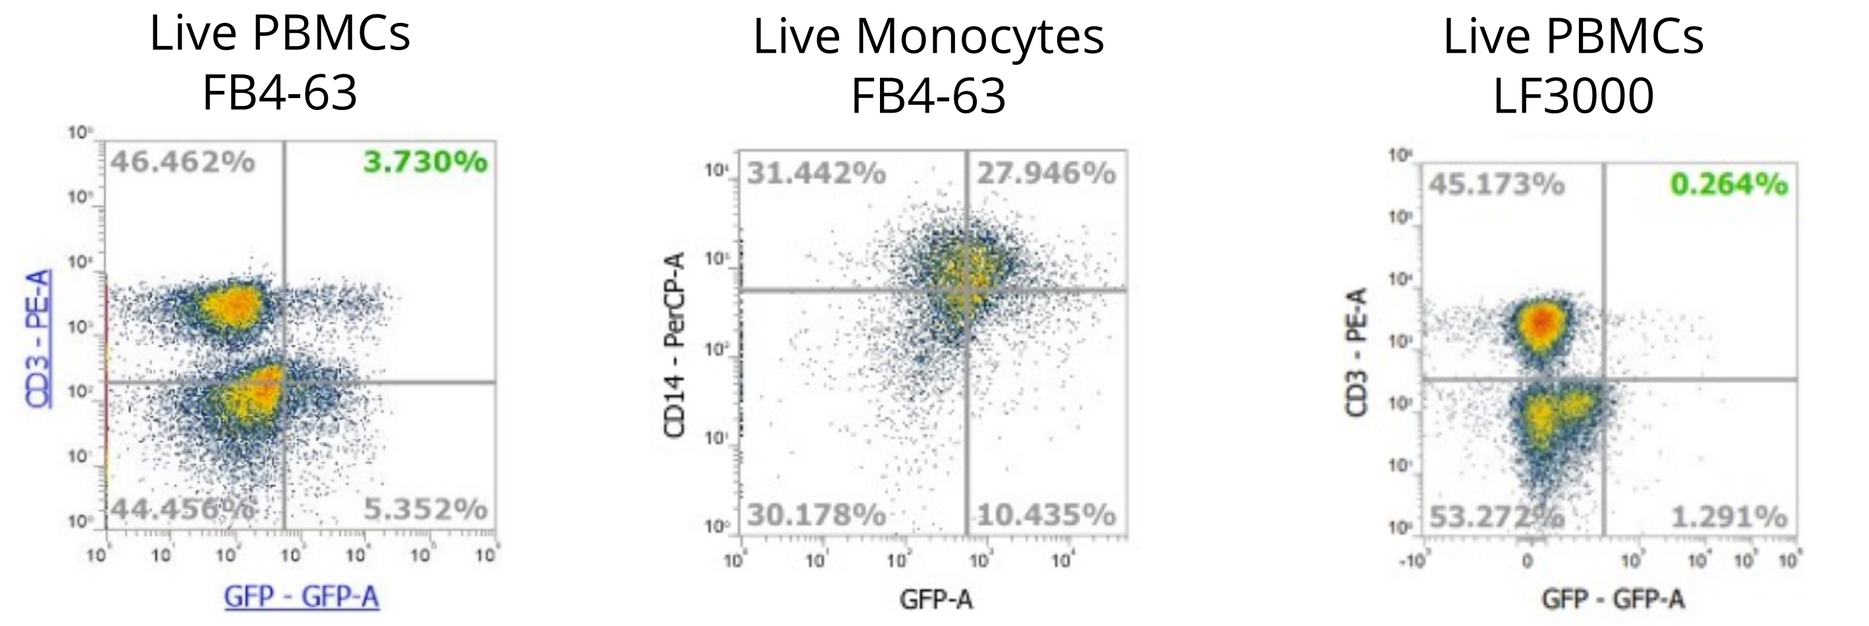
Live PBMCs FB4-63
46.462%
3.730%
44.456%
5.352%
Live Monocytes FB4-63
31.442%
27.946%
30.178%
10.435%
Live PBMCs LF3000
45.173%
0.264%
53.272%
1.291%

Figure 5. GFP expression in PBMCs treated with FB4-63 lipoplexes compared to Lipofectamine 3000.

4 Activated vs. Non-Activated PBMCs

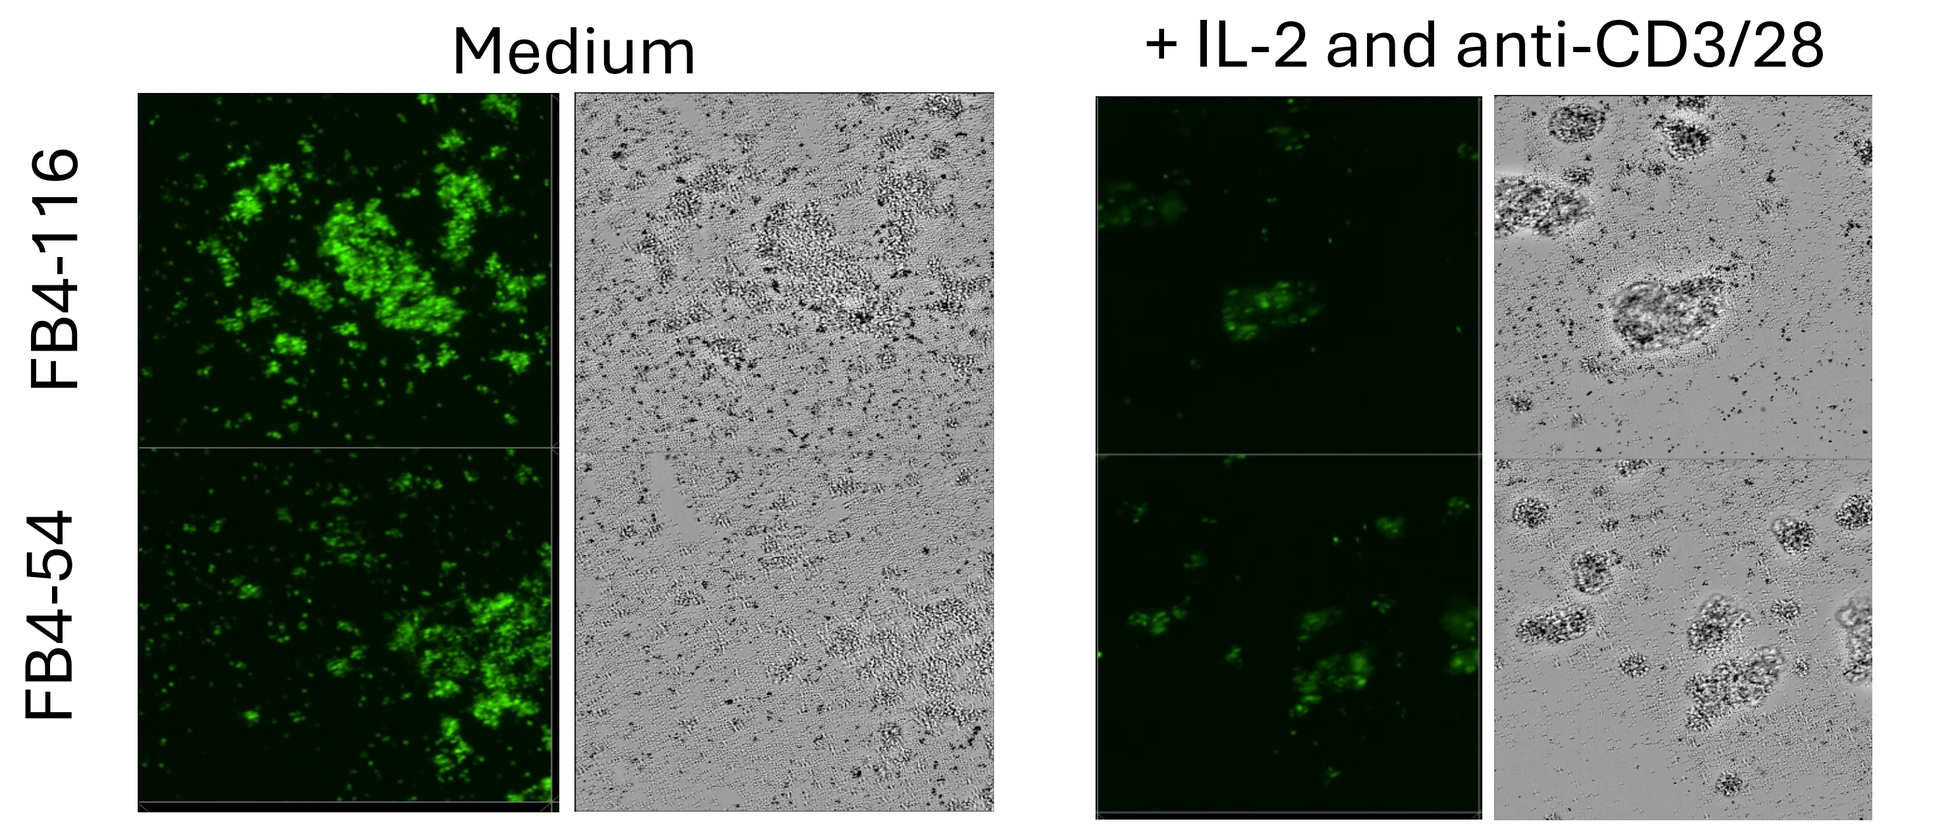
Medium
+ IL-2 and anti-CD3/28
FB4-116
FB4-54

Figure 6. Lipoplexes in PBMCs with and without T cell activators. 100,000 PBMCs were plated per well and incubated for 3 days in RPMI 1640+10% FBS. Half of the cells were cultured in media supplemented with T cell activators IL-2 and anti-CD3/28 until transfection. Media was replaced with fresh media containing 0.2ug of GFP-RNA complexed with an ionizable lipid. Cell images were taken from the 24 hr timepoint.

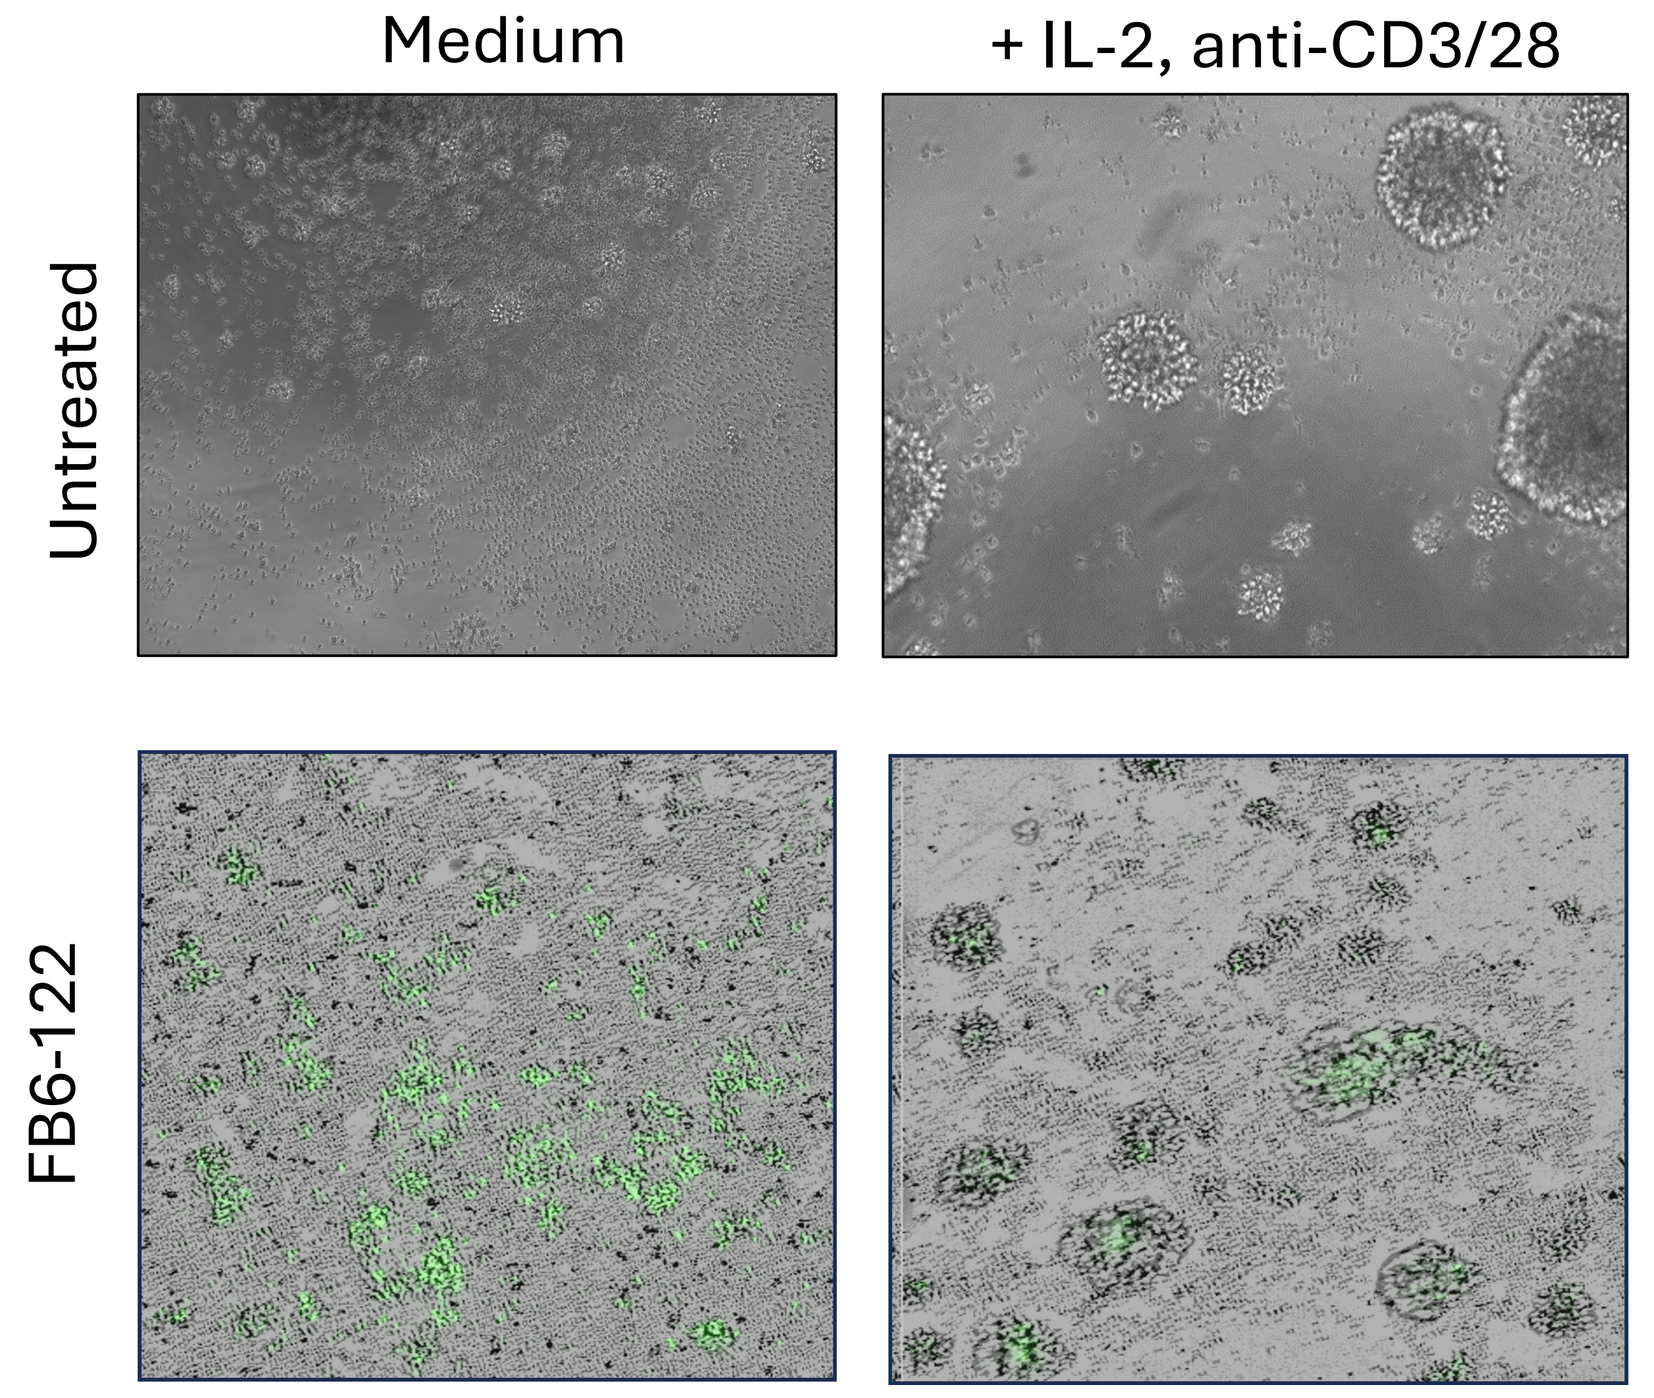
Medium
+ IL-2, anti-CD3/28
Untreated
FB6-122

Figure 7. FB6-122 lipoplex transfection of fresh human PBMCs. EGFP and brightfield image overlays show strong GFP expression in T cell aggregates in media supplemented with activators compared to GFP expression in non-activated and untreated cells. Cells were imaged 18hrs after transfection.

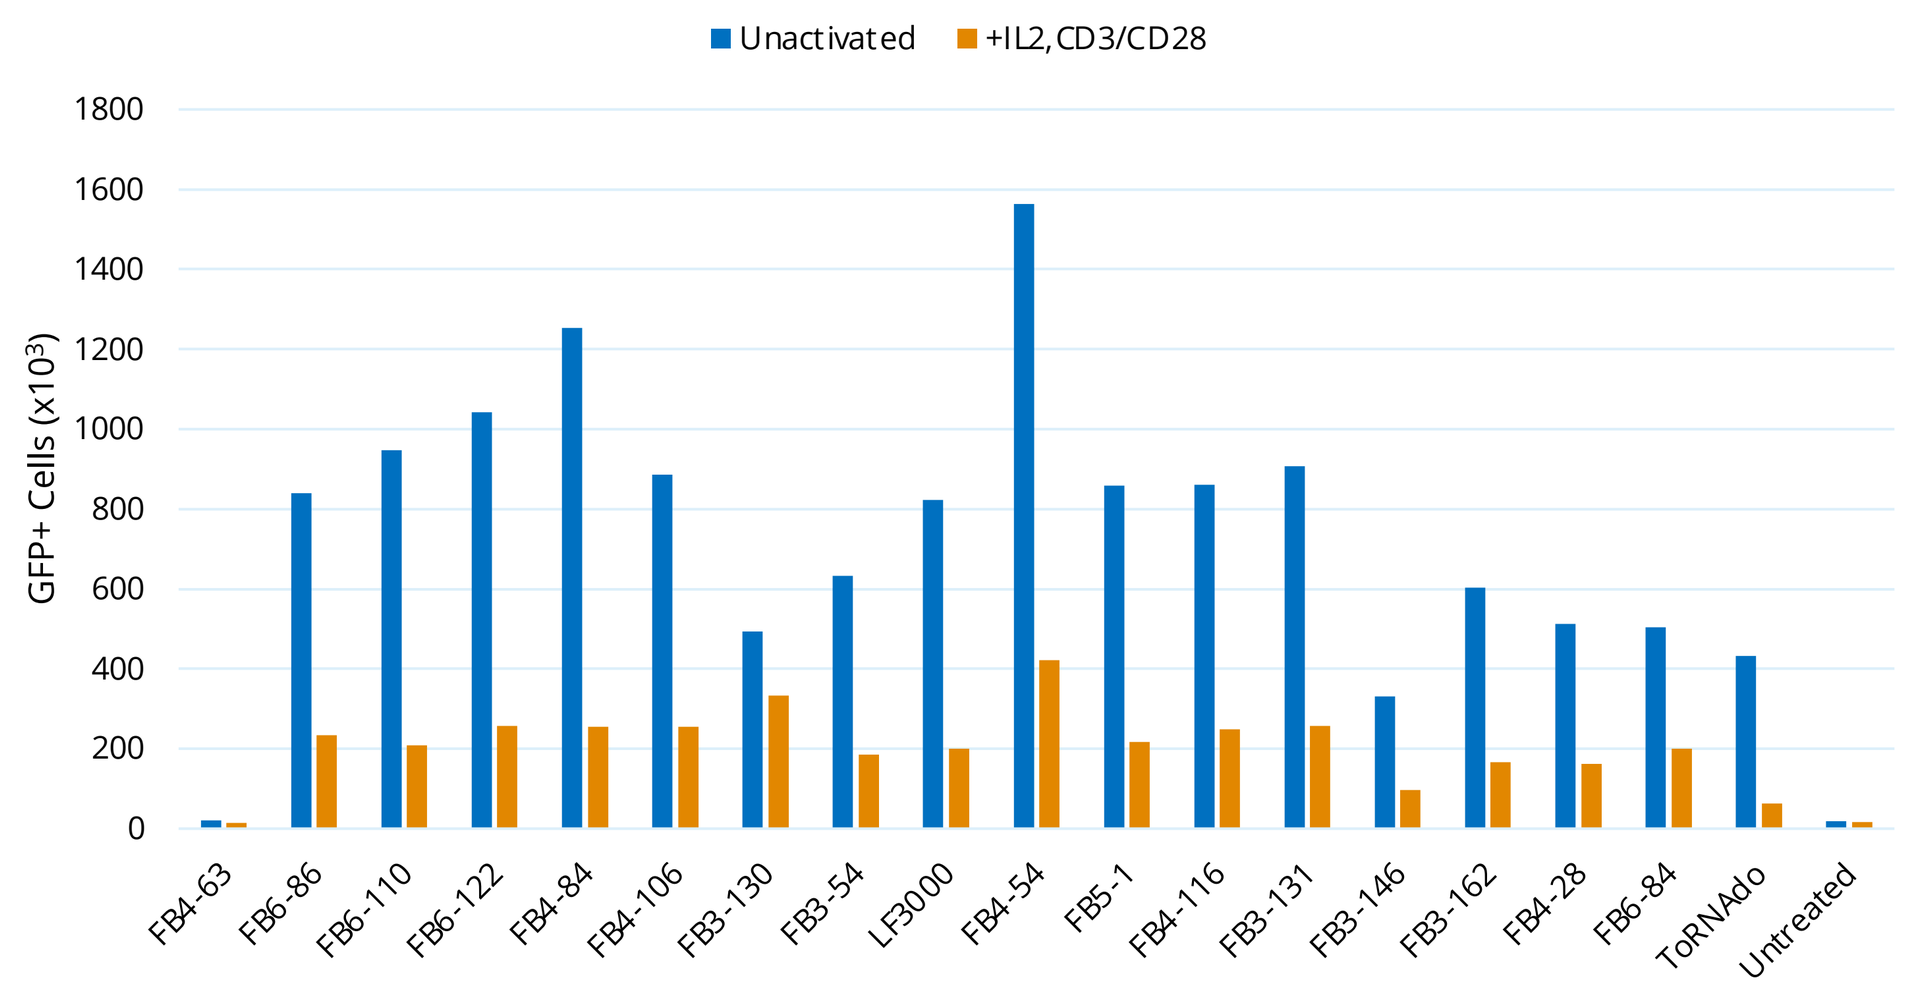
Unactivated
+IL2, CD3/CD28
GFP+ Cells (x10³)

Figure 8. GFP expression in PBMC lipid library screen. 17 structurally diverse ionizable lipids which had successfully transfected iMSCs were selected for transfecting 0.2ug RNA per 100,000 PBMCs in culture media with and without T cell activators. Several lipids yielded more GFP+ cells than Lipofectamine 3000.

5 THP-1 Monocyte Transfections

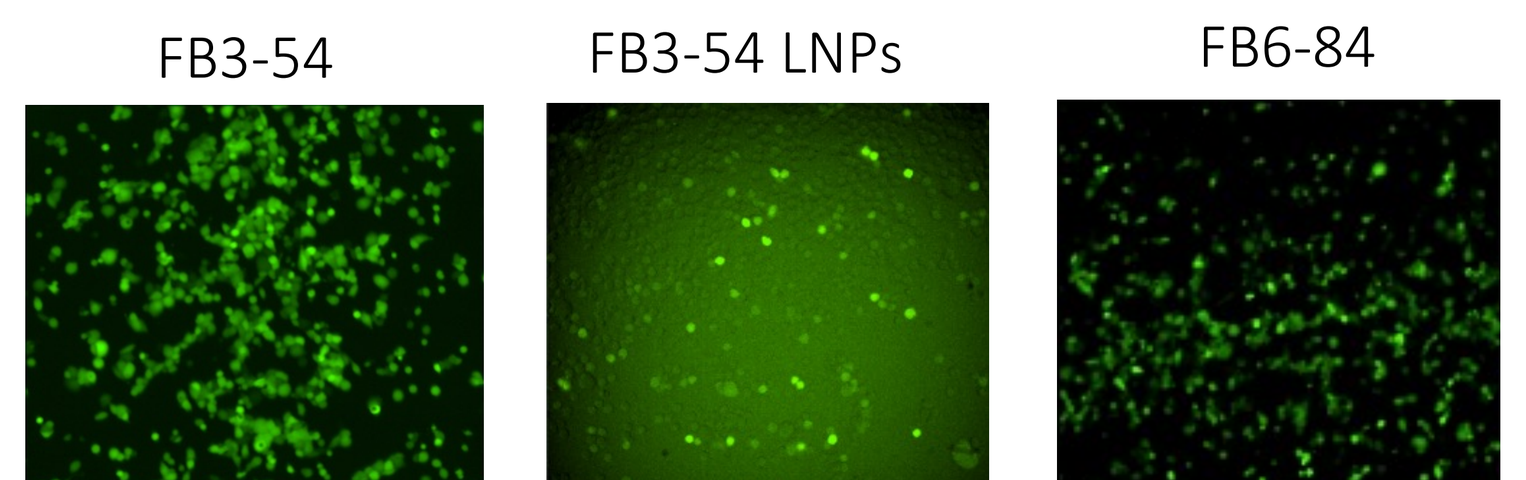
FB3-54
FB3-54 LNPs
FB6-84

Figure 9. FB3-54 and FB6-84 lipoplexes and FB3-54 LNPs successfully transfected THP-1 cells. 0.1ug of mRNA was delivered via lipoplexes and 62.5ng via LNPs per 10,000 cells. GFP expression was imaged at 24 hrs.

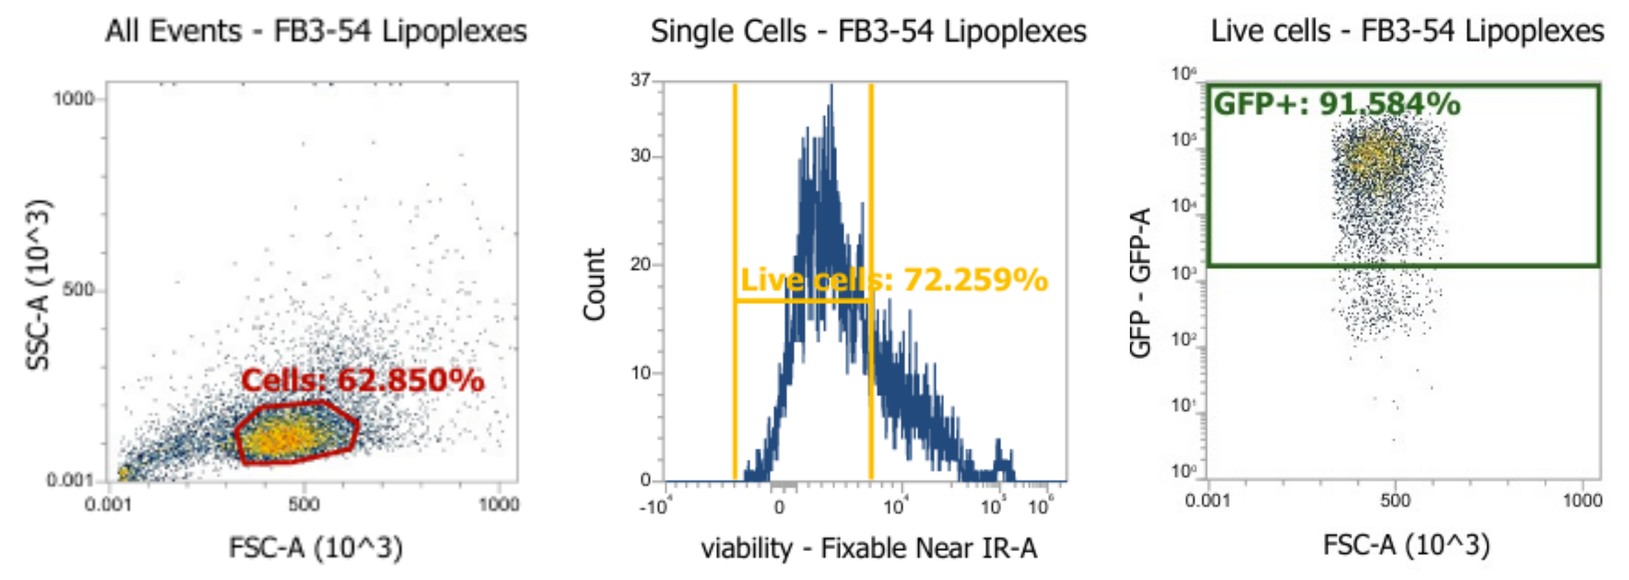
All Events - FB3-54 Lipoplexes
Cells: 62.850%
Single Cells - FB3-54 Lipoplexes
Live cells: 72.259%
Live cells - FB3-54 Lipoplexes
GFP+: 91.584%

Figure 10. Flow cytometry of THP-1 cells transfected with FB3-54 lipoplexes at a 2:1 ratio. FB3-54 lipoplexes yielded over 91% transfection efficiency and cell viability over 72%.

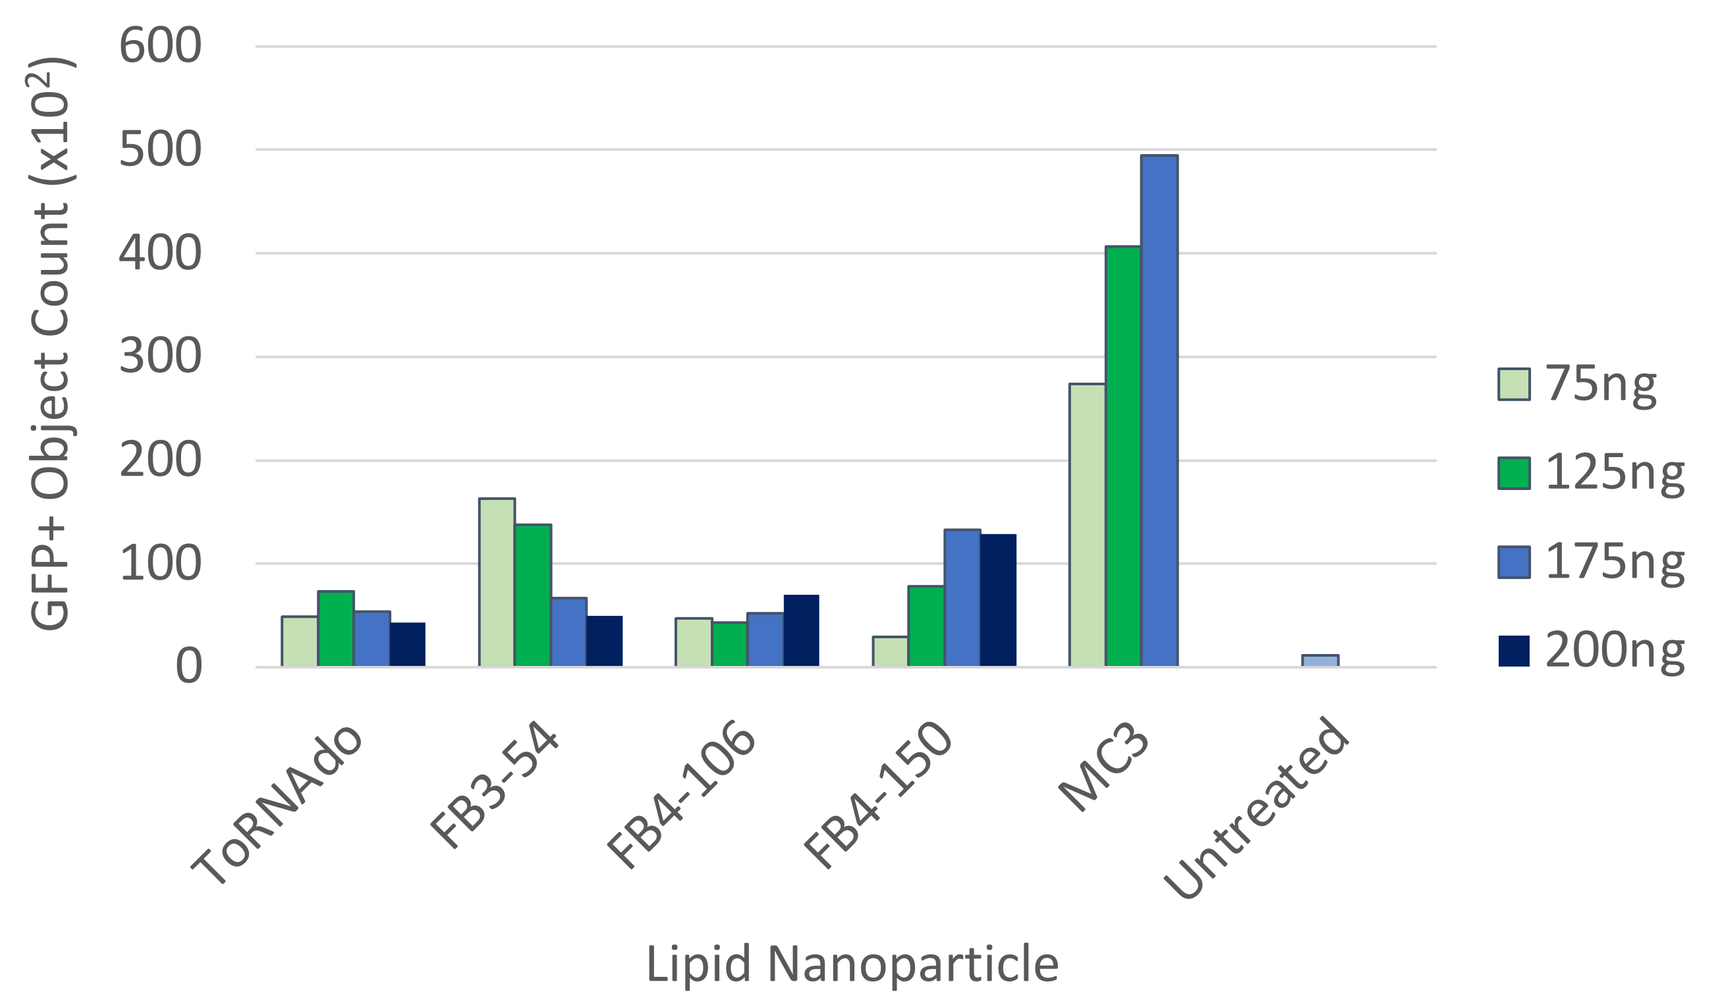
GFP+ Object Count (x10²)
75ng
125ng
175ng
200ng
ToRNAdo
FB3-54
FB4-106
FB4-150
MC3
Untreated
Lipid Nanoparticle

Figure 11. LNP transfection efficiency in THP-1 cells measured by flow cytometry. Optimal RNA dose varied greatly among LNP types between 75ng and 200ng per 10,000 cells. Among the four novel lipids tested, FB3-54 LNPs yielded the highest GFP expression at the lowest dose. MC3 LNPs performed best overall with highest GFP signal at 200ng. Flow cytometry was conducted at 24 hrs.

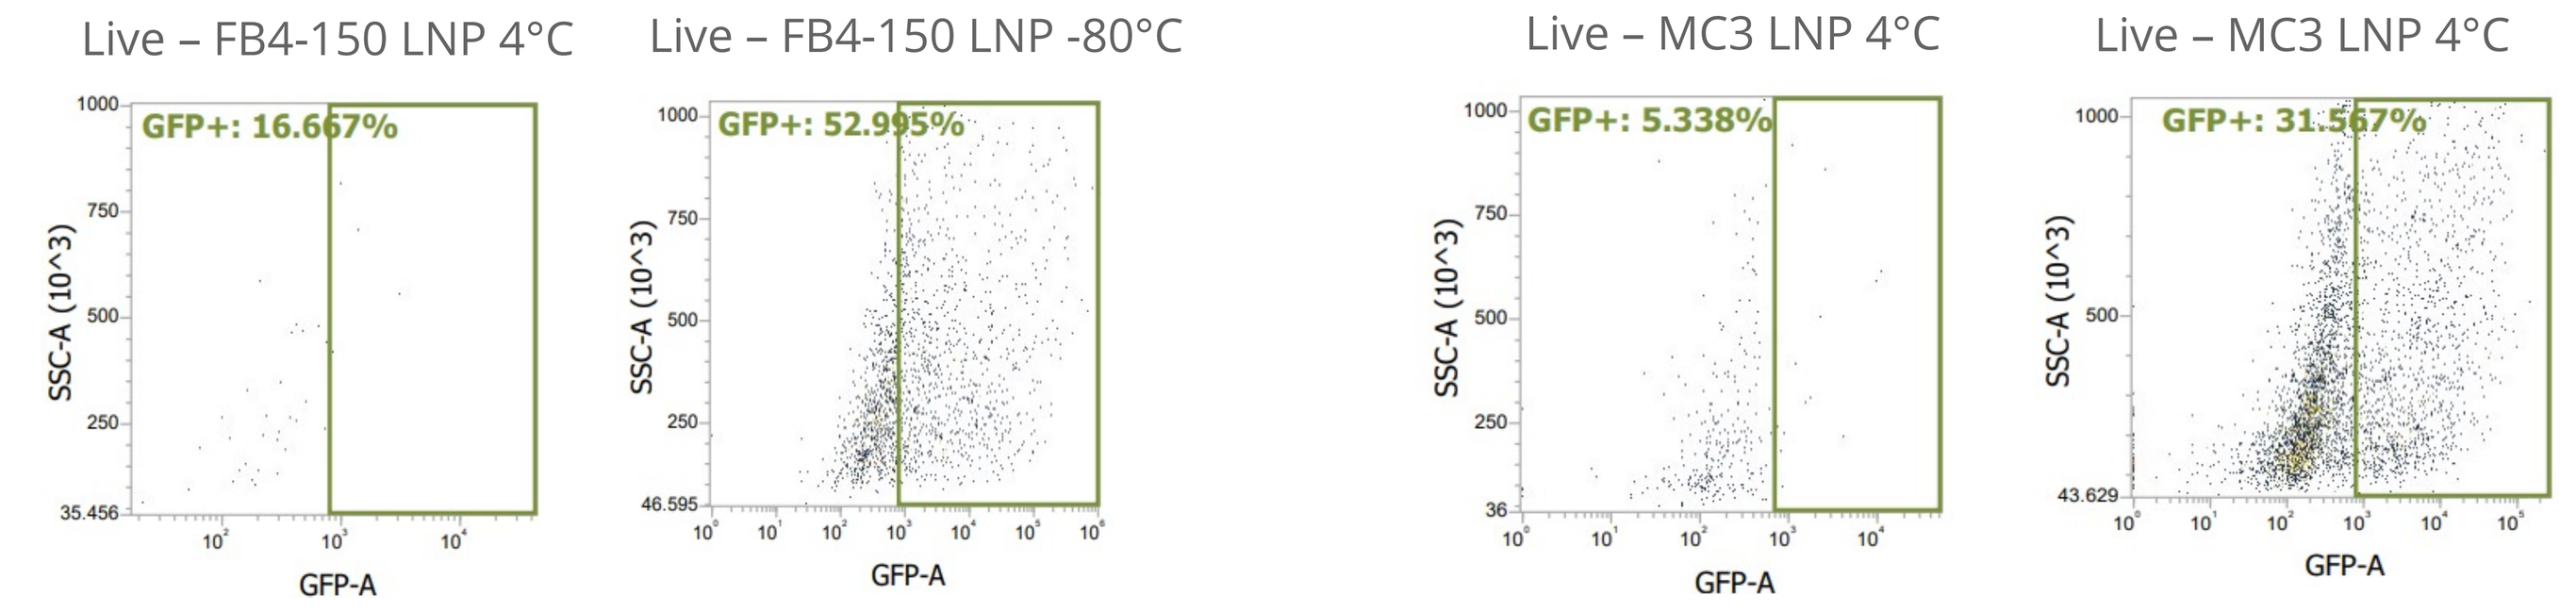
Live – FB4-150 LNP 4°C
GFP+: 16.667%
Live – FB4-150 LNP -80°C
GFP+: 52.995%
Live – MC3 LNP 4°C
GFP+: 5.338%
Live – MC3 LNP 4°C
GFP+: 31.567%

Figure 12. LNP storage temperature affects GFP expression in transfected THP-1 cells. To study the effect of storage temperature on LNP transfection efficiency, prepared LNPs were stored at temperatures of 4°C or snap-frozen in LN2 and thereafter stored at 80°C. After storing for 1 week, LNPs were thawed at room temperature and diluted into THP-1 culture medium. 0.5mL treatment volumes were delivered per well (400,000 cells/well). GFP expression of the live cell population was assessed 24 hrs after transfection by flow cytometry. FB4-150 and MC3 LNPs yielded 96.1% and 98.5% viability, respectively. Both lipids produced over 3-fold higher GFP expression when snap-frozen and stored at 80°C for a week compared to the 4°C condition.

6 iPSCs and iPSC-Derived Cell Transfections

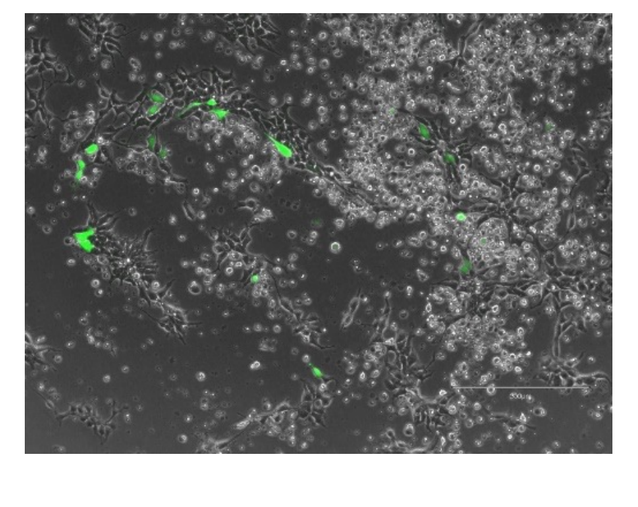

Figure 13. LNPs deliver mRNA into iPSCs. 480ng RNA was delivered per 40,000 iPSCs via ToRNAdo LNPs stored at -80°C for one week. GFP expression peaked at 18 hrs.

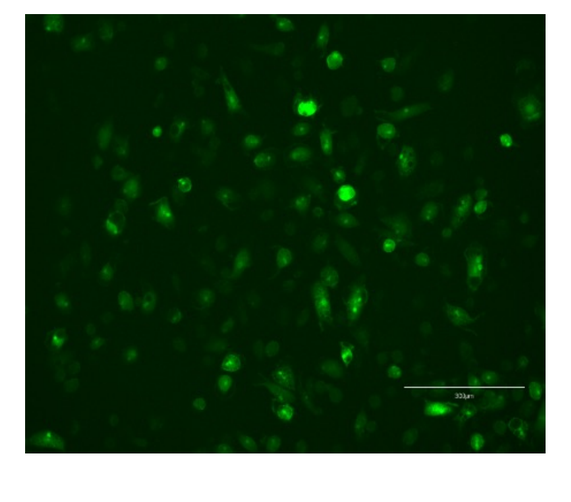

Figure 14. ToRNAdo lipoplexes in iPSC-derived macrophages. 0.1ug RNA was delivered per 50,000 cells. Peak GFP occurred at 24 hrs.

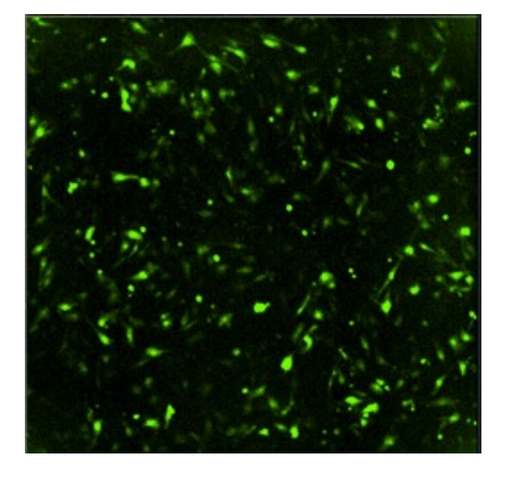

Figure 15. FB6-146 lipoplexes in iMSCs. Peak GFP occurred at 14 hrs for 0.1ug RNA per 50,000 cells.

American Society of Gene + Cell Therapy