FACTOR BIOSCIENCE

Chromatin Opening Elements Mitigate Silencing of Transgenes During iPSC to Macrophage or Lymphocyte Differentiation

Claire Aibel[1], Abigail Blatchford[1], Christopher B. Rohde[1], Matthew Angel[1]
[1]Factor Bioscience Inc., Cambridge, MA
This work is protected by one or more pending patent applications.

Summary

Differentiation of gene-edited induced pluripotent stem cells (iPSCs) holds potential for superior production of genetically identical transgene-expressing immune cells that can be used for therapeutic applications. However, transgenes driven by synthetic promoters can become silenced during differentiation. Here, we demonstrate that insertion of a novel upstream regulatory sequence reduces differentiation-induced transgene silencing during iPSC to macrophage or lymphocyte differentiation.

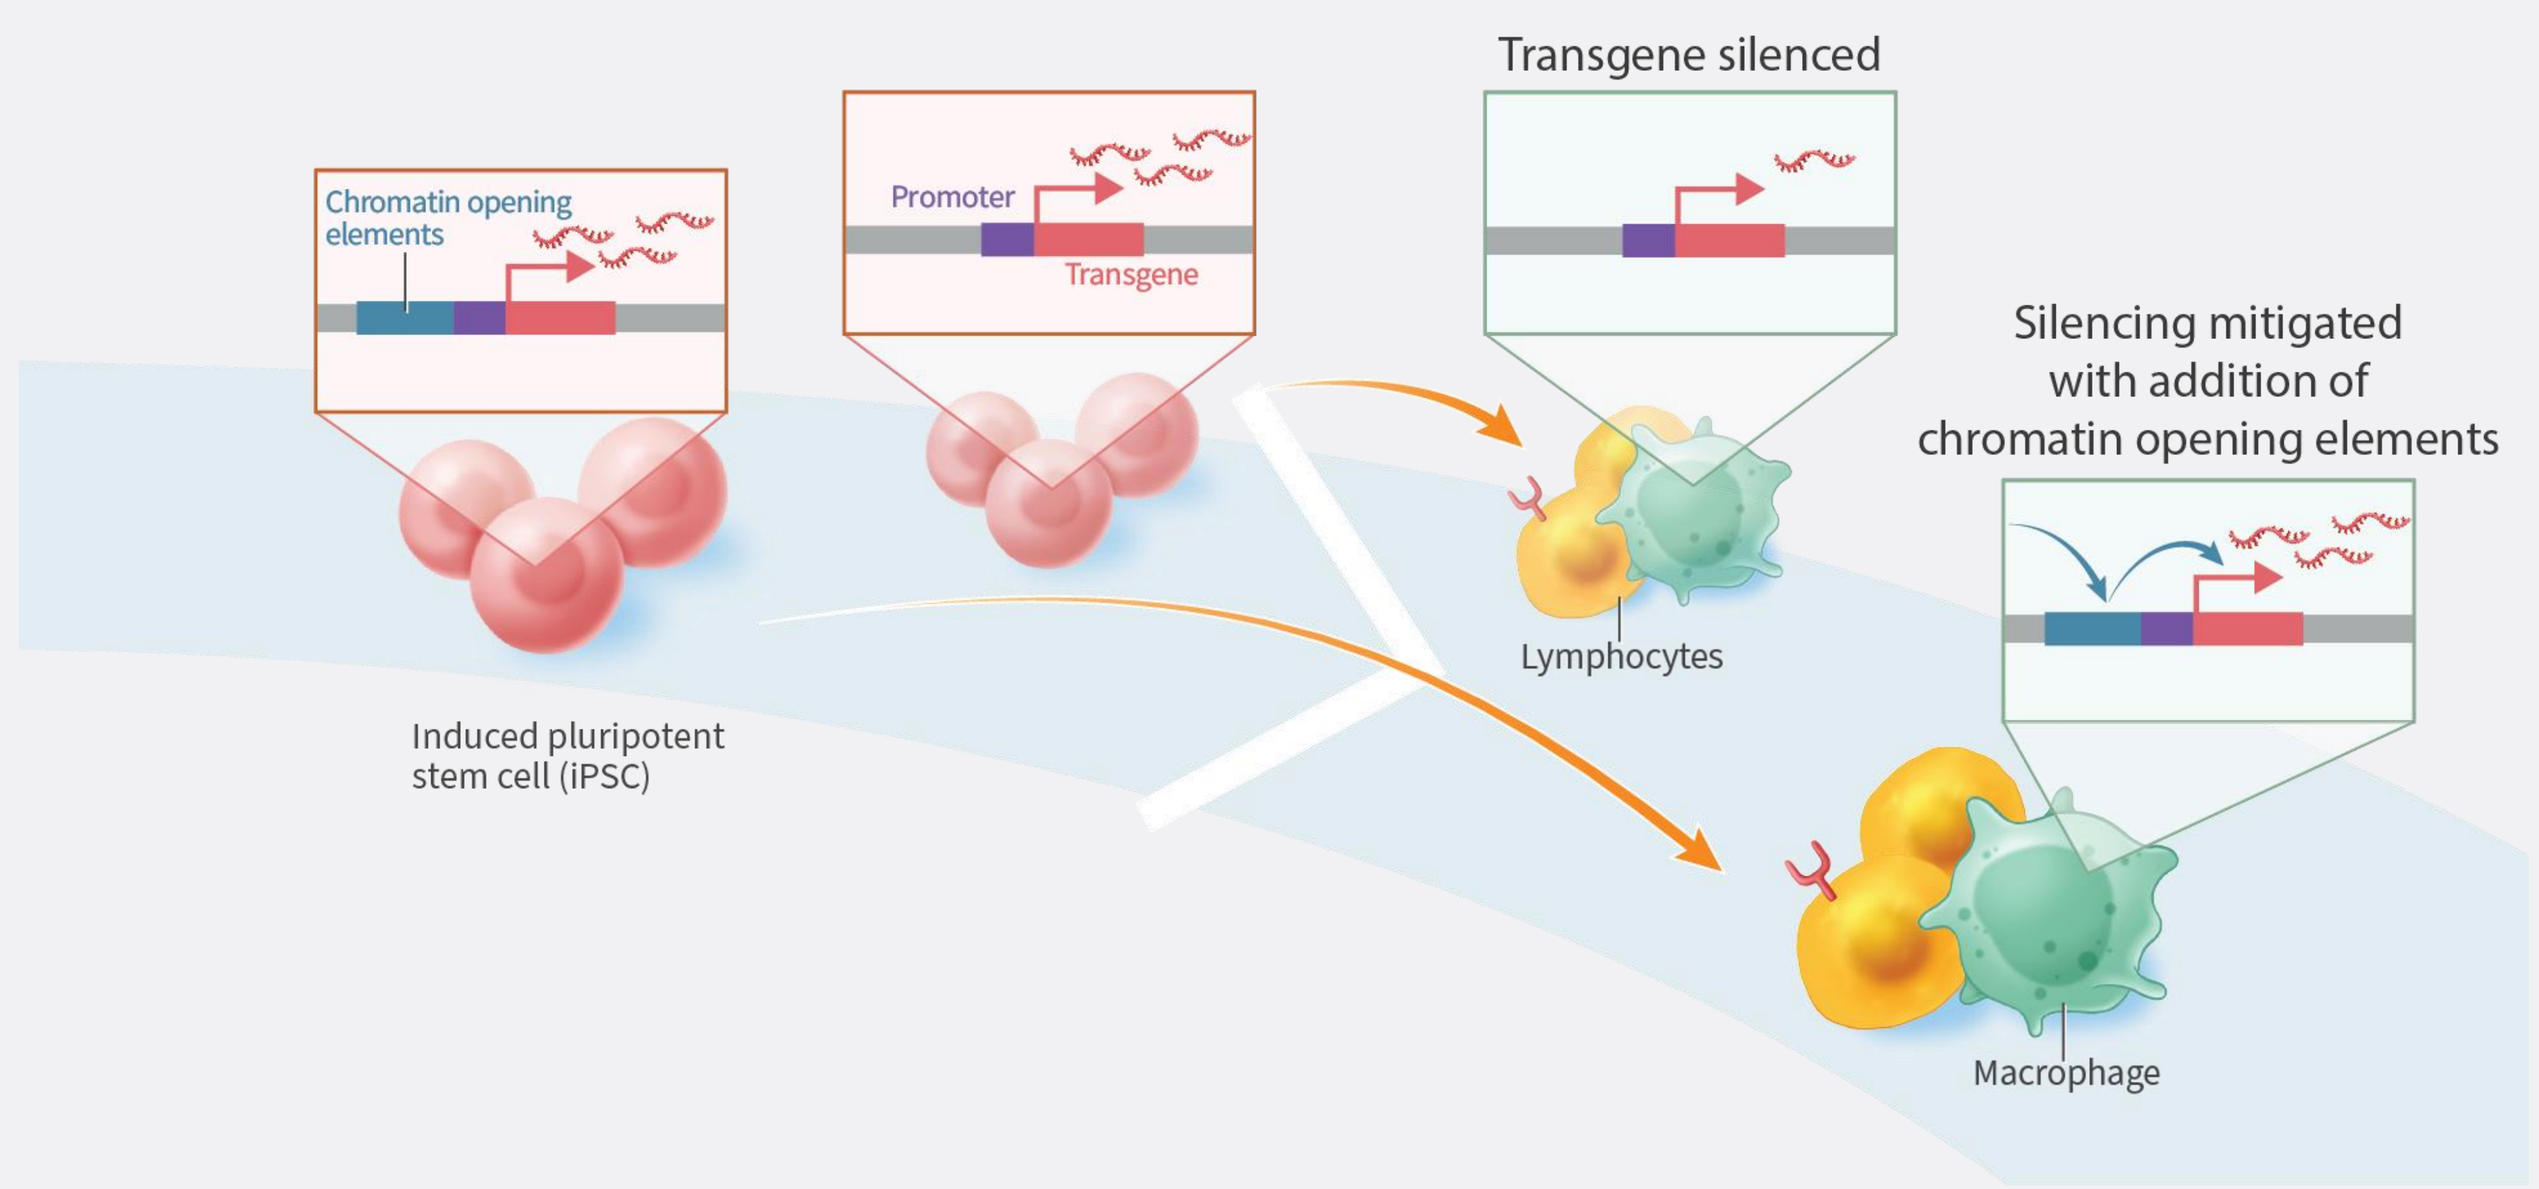
Chromatin opening elements
Promoter
Transgene
Transgene silenced
Silencing mitigated with addition of chromatin opening elements
Lymphocytes
Induced pluripotent stem cell (iPSC)
Macrophage

Conclusions

Here, we demonstrate that incorporation of a novel regulatory sequence confers a 9-fold or 40-fold increase in the percentage of transgene-expressing iPSC-derived macrophages and iPSC-derived lymphocytes, respectively. Furthermore, transgenic iPSCs and iPSC-derived cells display comparable key surface marker expression to wild-type cells. Therefore, these sequence elements may prove useful in engineering cells with functional transgenes encoding therapeutic proteins, which could contribute to the development of effective iPSC-based therapies.

1 Development and assessment of gene-edited iPSCs

Development of iPS cell lines containing transgenes with enhanced regulatory features

Figure 1. Development of gene-edited iPS cells. Figure 1a. Donor single-stranded DNA consisting of an ~800 base long universal chromatin opening element (UCOE) with 73% GC content, a synthetic promoter (EF1α or SFC), a GFP reporter, and AAVS1 homology arms were synthesized. Figure 1b. The donor DNA was electroporated into iPSCs at the AAVS1 safe-harbor locus. iPSCs were then singularly deposited into 96-well plates and harvested after sufficient colony growth. Single colonies were evaluated via PCR and those containing biallelic transgene insertions were expanded and used for downstream differentiations.

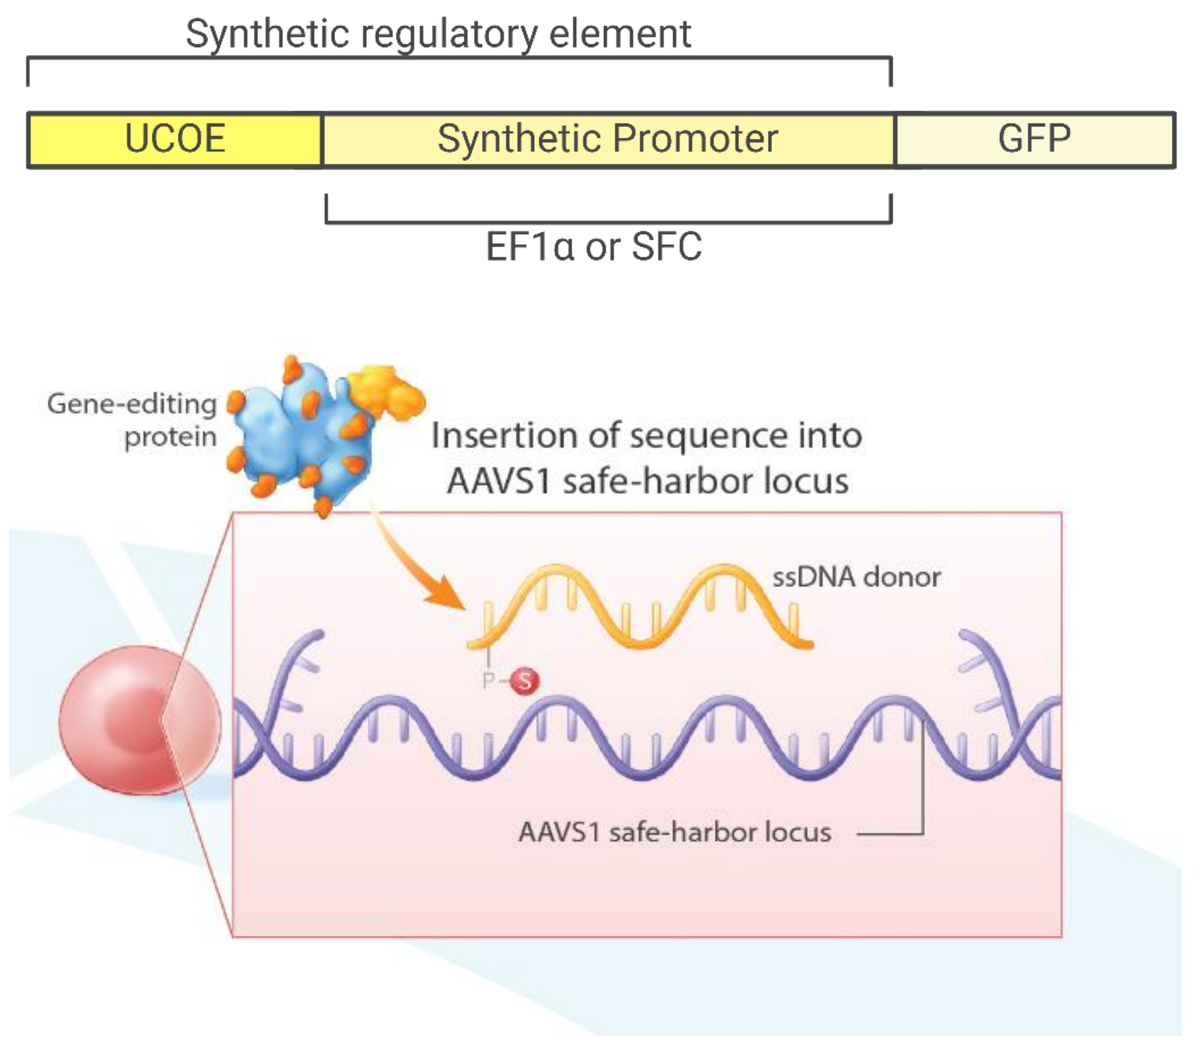
(1a)
Synthetic regulatory element
UCOE
Synthetic Promoter
GFP
EF1α or SFC
(1b)
Gene-editing protein
Insertion of sequence into AAVS1 safe-harbor locus
ssDNA donor
AAVS1 safe-harbor locus

Regulatory sequence-dependent transgene expression

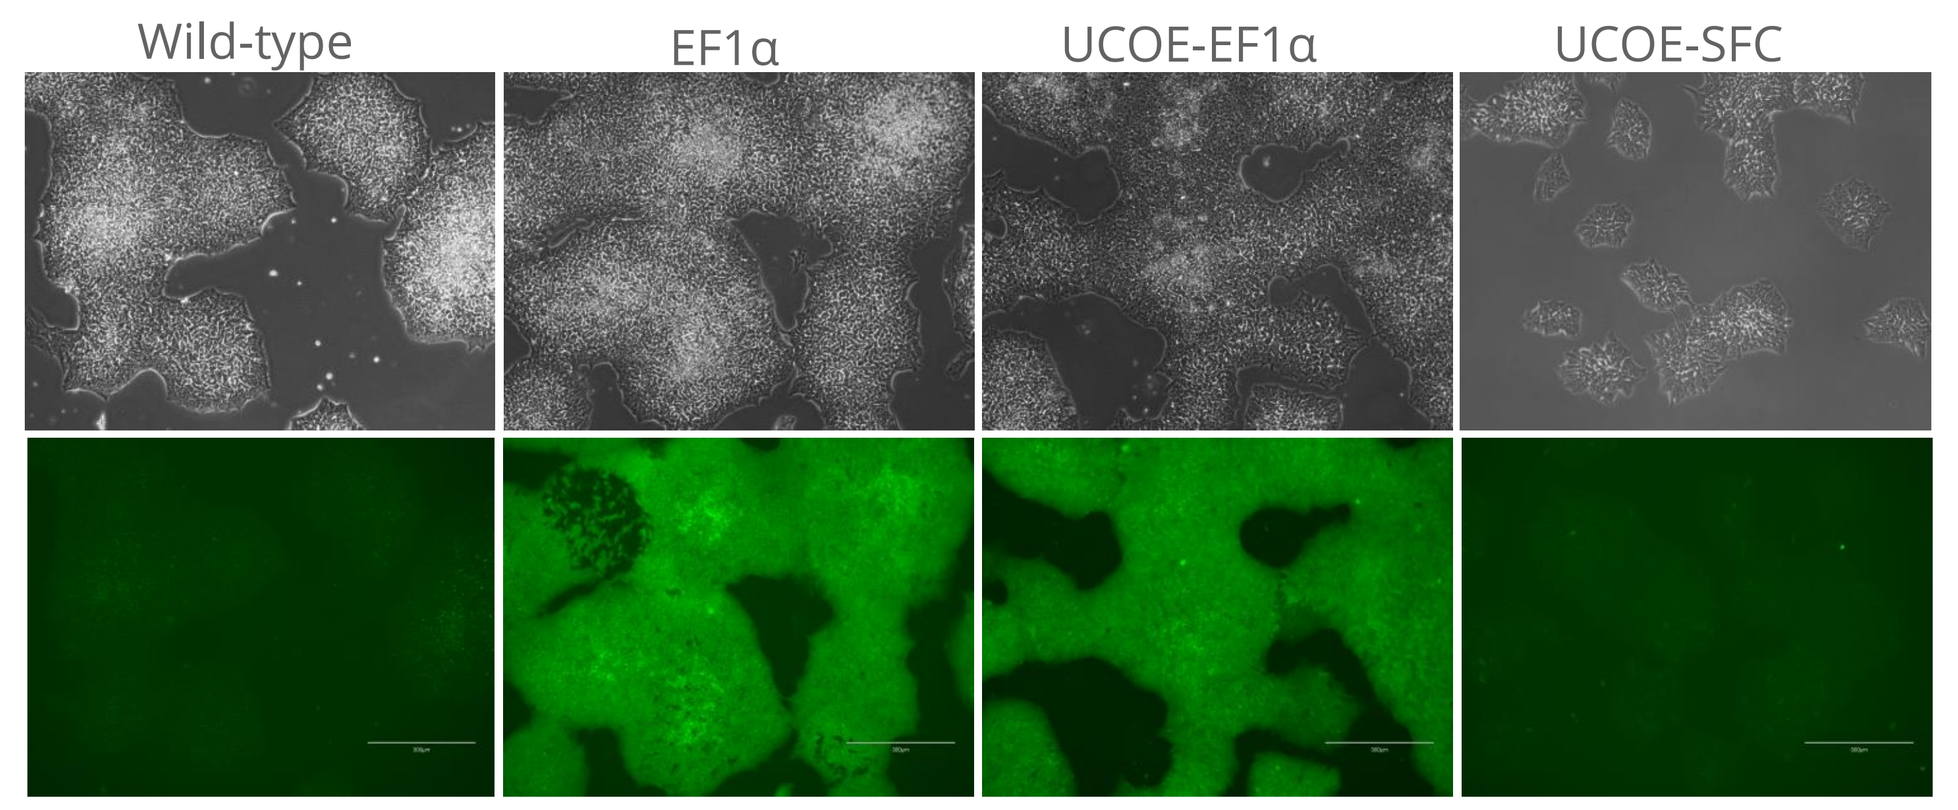
(2a)
Wild-type
EF1α
UCOE-EF1α
UCOE-SFC

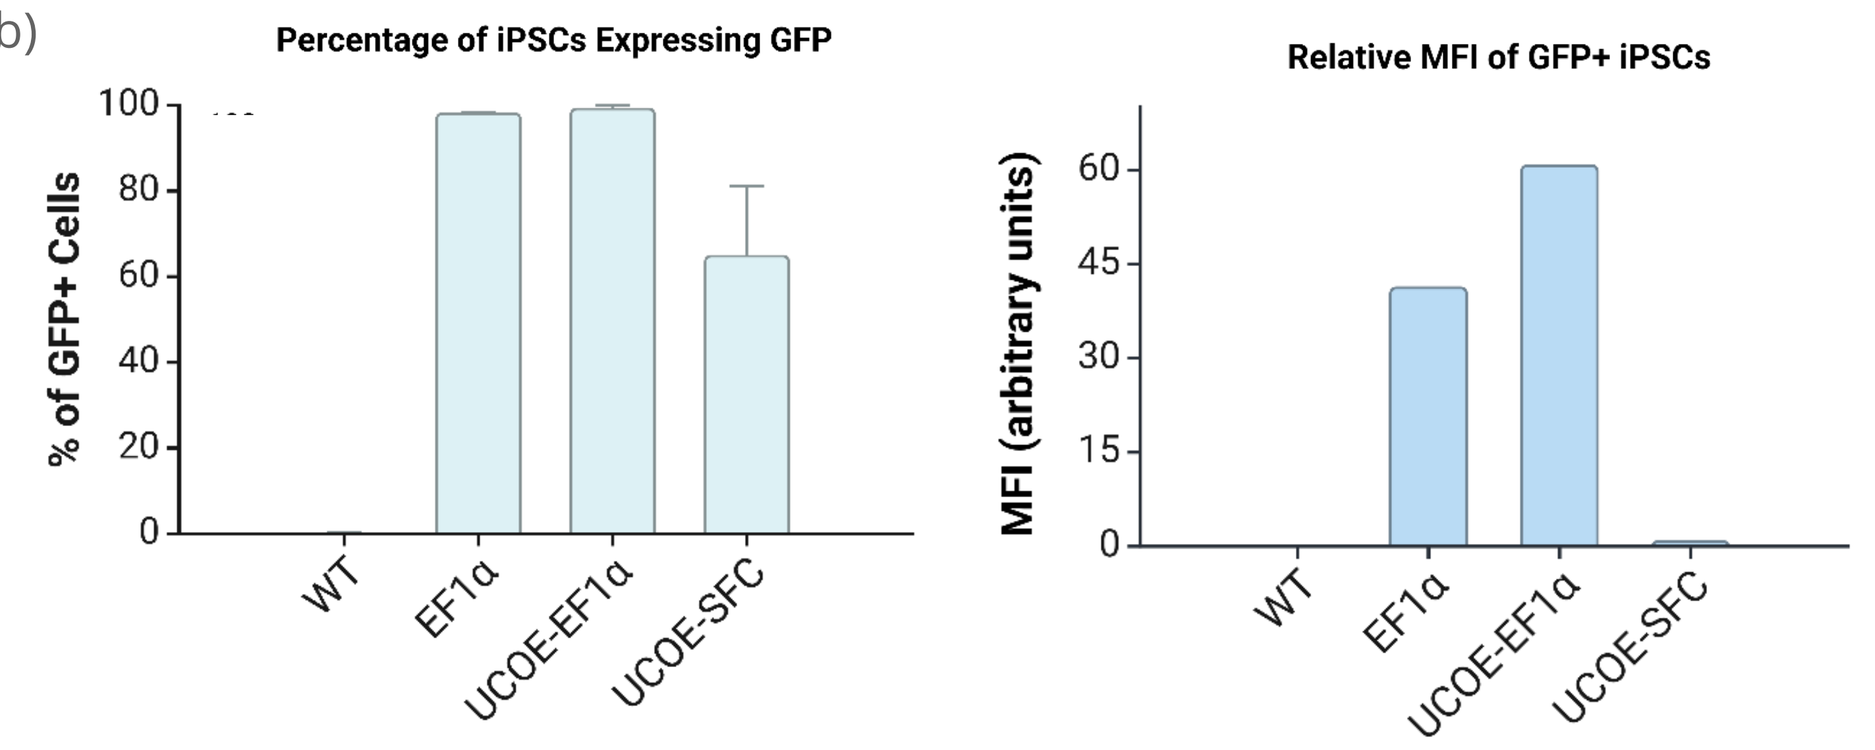
(2b)
Percentage of iPSCs Expressing GFP
% of GFP+ Cells
100
80
60
40
20
0
WT
EF1α
UCOE-EF1α
UCOE-SFC
Relative MFI of GFP+ iPSCs
MFI (arbitrary units)
60
45
30
15
0
WT
EF1α
UCOE-EF1α
UCOE-SFC

Figure 2. GFP expression in undifferentiated gene-edited iPS cell lines. Figure 2a. Brightfield (top) and GFP (bottom) images of wild-type and gene-edited iPSCs prior to differentiation. Figure 2b. Left: Percentage of GFP+ iPSCs determined using flow cytometry (n=3). Error bars represent SEM. Right: Relative GFP median fluorescence intensity (MFI), determined using flow cytometry, among the GFP+ iPSCs.

Assessment of key pluripotency markers

Figure 3. Assessment of pluripotency markers in undifferentiated iPSCs. Figure 3a. Percentage of iPSCs expressing pluripotency markers TRA-1-60 and TRA-1-81 determined using flow cytometry. Figure 3b. Representative density plots acquired using flow cytometry to illustrate gating strategy used to determine percentage of TRA-1-60+ cells in the UCOE-EF1α iPSCs.

(3a)

Surface marker	Expected	Wild-type	EF1α	UCOE-EF1α	UCOE-SFC
TRA-1-60	+	96.9%	95.2%	99.9%	99.9%
TRA-1-81	+	97.0%	94.7%	99.9%	99.9%

(3b)

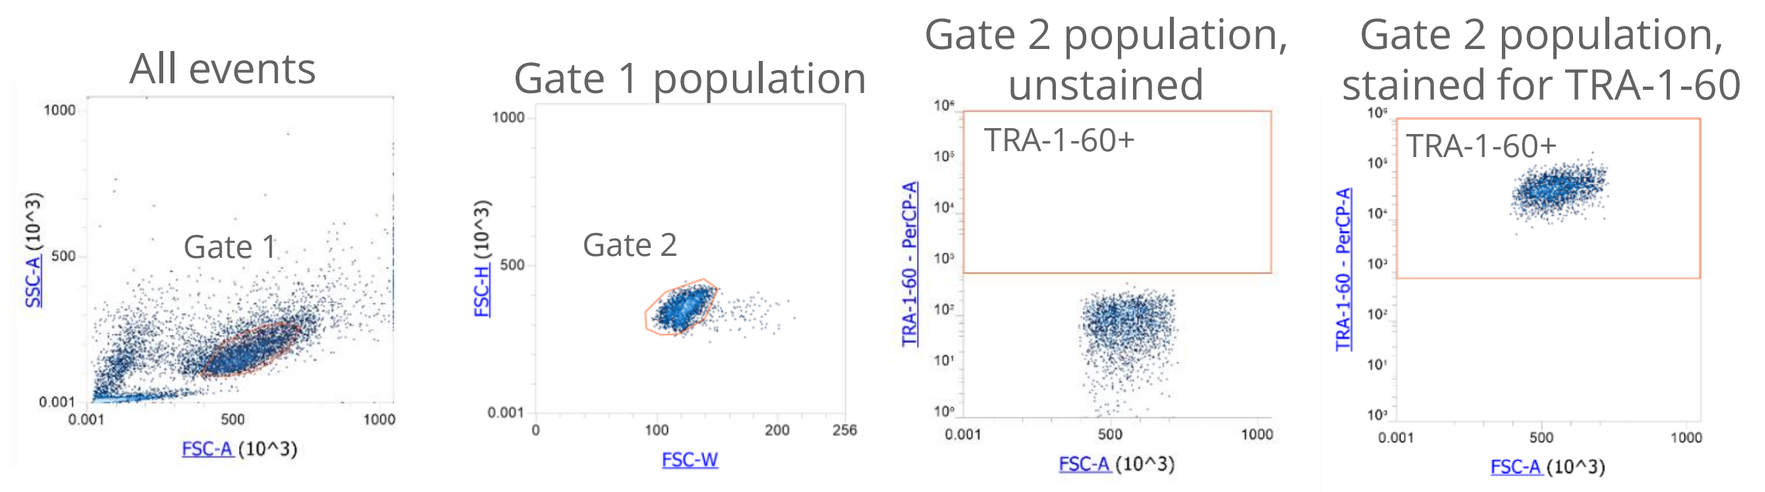
All events
Gate 1
Gate 1 population
Gate 2
Gate 2 population, unstained
TRA-1-60+
Gate 2 population, stained for TRA-1-60
TRA-1-60+

2 Characterization of transgenic iPSC-derived macrophages

iPSC → iMacrophage differentiation overview

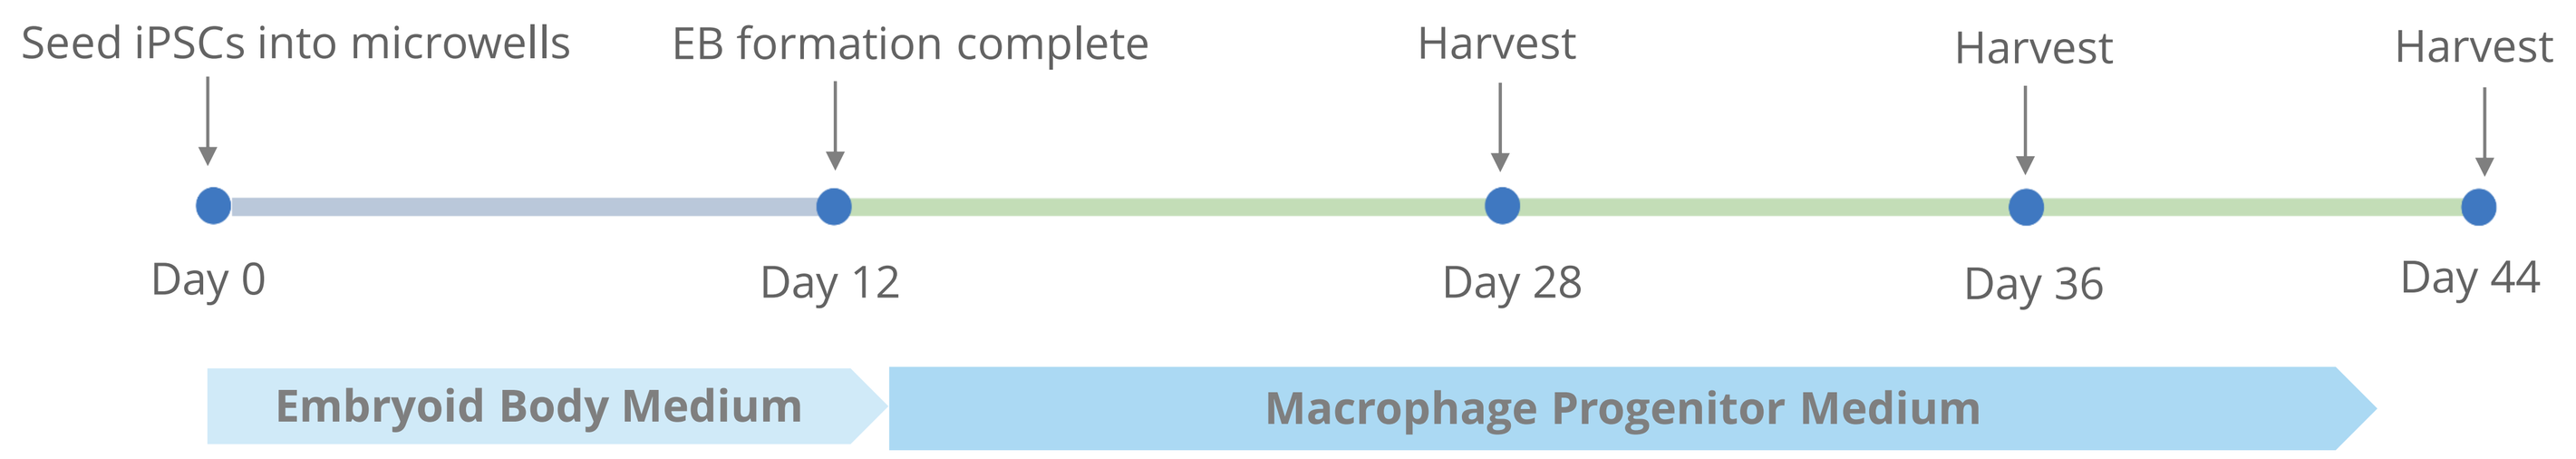
Seed iPSCs into microwells
EB formation complete
Harvest
Harvest
Harvest
Day 0
Day 12
Day 28
Day 36
Day 44
Embryoid Body Medium
Macrophage Progenitor Medium

Figure 4. Differentiation of iPSCs into macrophages. Gene-edited iPSCs were seeded into microwells on day 0 in Embryoid Body (EB) Formation Medium (STEMCELL Technologies) and transferred to a non-tissue culture treated plate on day 5. On day 12, EBs were transferred to macrophage progenitor expansion medium in Matrigel®-coated cultureware. Macrophages developed from adherent EBs and were evaluated via flow cytometry on days 28, 36, and 44.

iMacrophages with UCOEs display higher transgene expression

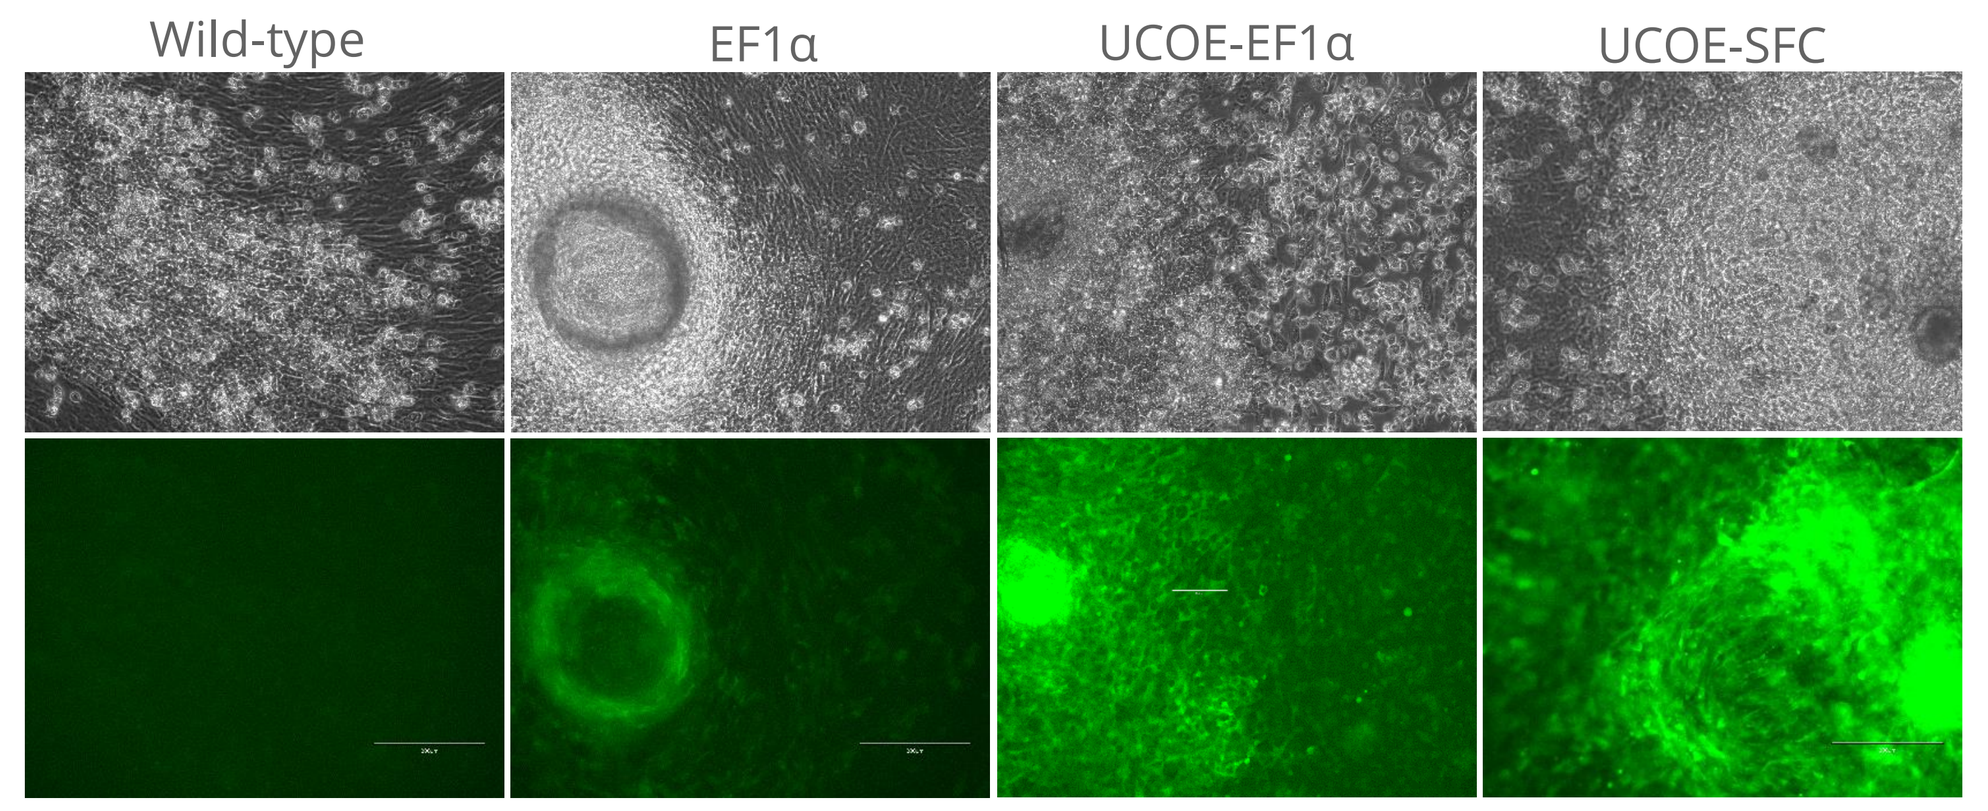
(5a)
Wild-type
EF1α
UCOE-EF1α
UCOE-SFC

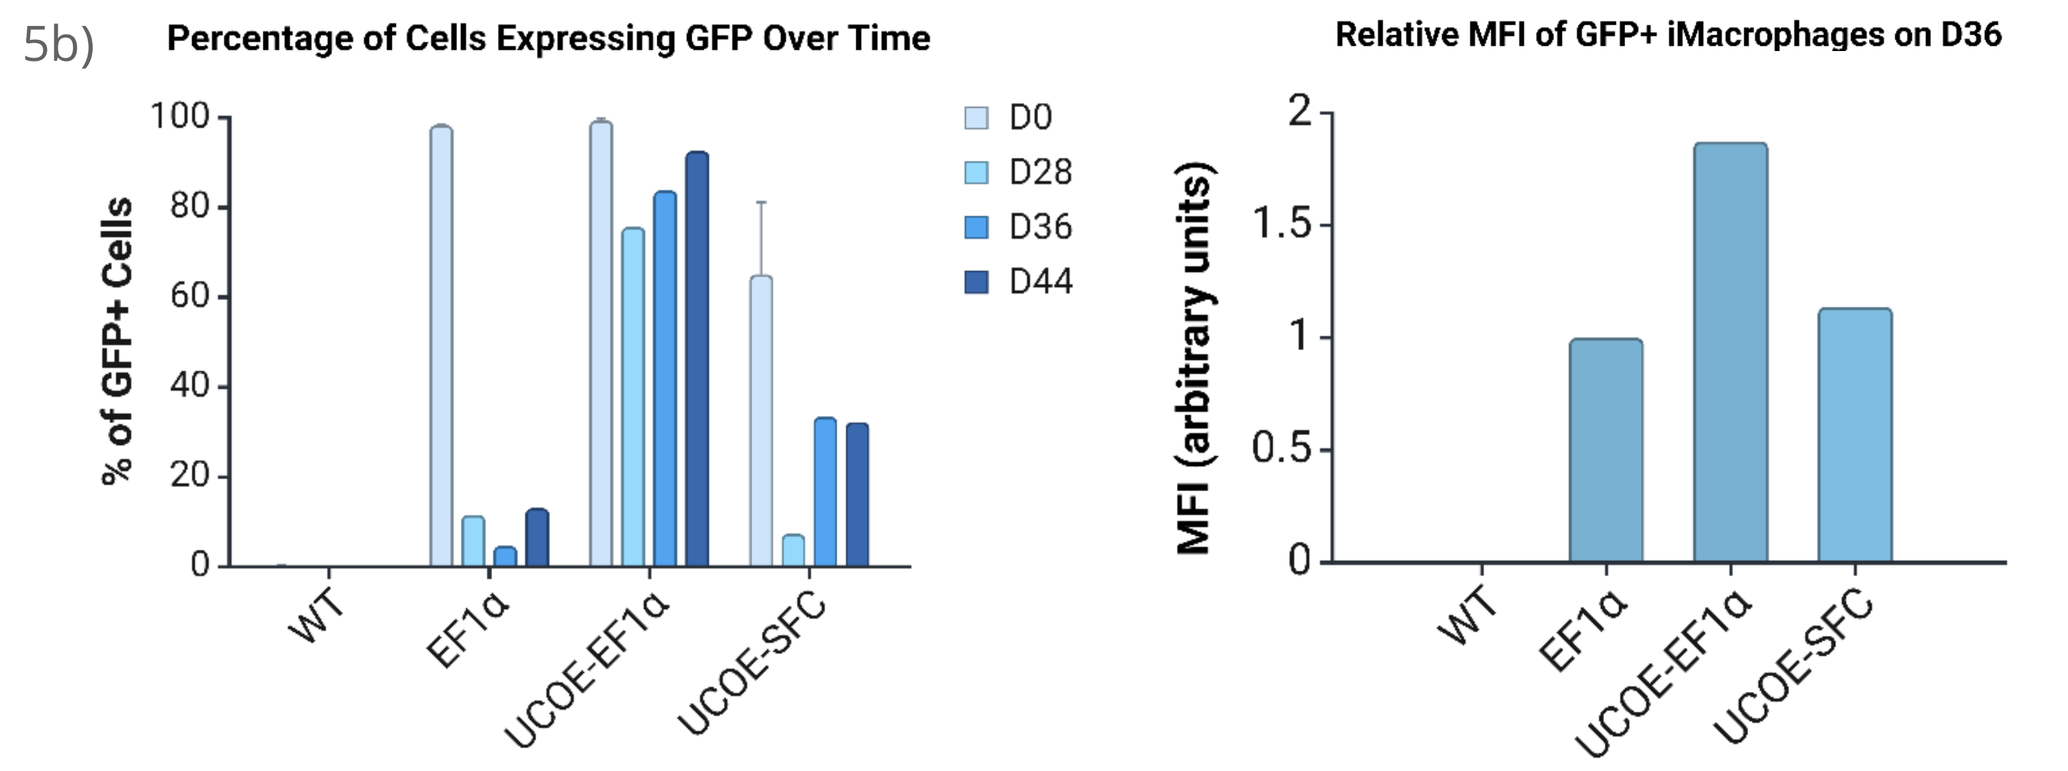
5b)
Percentage of Cells Expressing GFP Over Time
% of GFP+ Cells
D0
D28
D36
D44
WT
EF1α
UCOE-EF1α
UCOE-SFC
Relative MFI of GFP+ iMacrophages on D36
MFI (arbitrary units)
2
1.5
1
0.5
0

Figure 5. Transgene expression in iMacrophages on days 28, 36, and 44. Figure 5a. Brightfield (top) and GFP (bottom) images of iPSC-derived macrophages on day 36 of differentiation. Figure 5b. Left: Percentage of cells expressing GFP, assessed using flow cytometry, on days 0, 28, 36, and 44 of differentiation. Error bars represent SEM. Right: Relative median fluorescence intensity of GFP+ cells on day 36 of differentiation.

Assessment of key macrophage markers

Surface marker	Expected	Wild-type	EF1α	UCOE-EF1α	UCOE-SFC
CD14	+	87.7%	97.2%	99.6%	99.6%
CD45	+	99.6%	98.9%	99.6%	99.6%
CD64	+	98.9%	88.6%	96.7%	92.3%

Figure 6. Assessment of key macrophage markers on day 36 of differentiation. Expression of CD14 (lipopolysaccharide binding receptor), CD45 (leukocyte common antigen), and CD64 (Fc gamma receptor) were evaluated using flow cytometry. Both wild-type and gene-edited iPSC-derived macrophages were positive for CD14, CD45, and CD64.

3 Characterization of transgenic iPSC-derived lymphocytes

iPSC → iLymphocyte differentiation overview

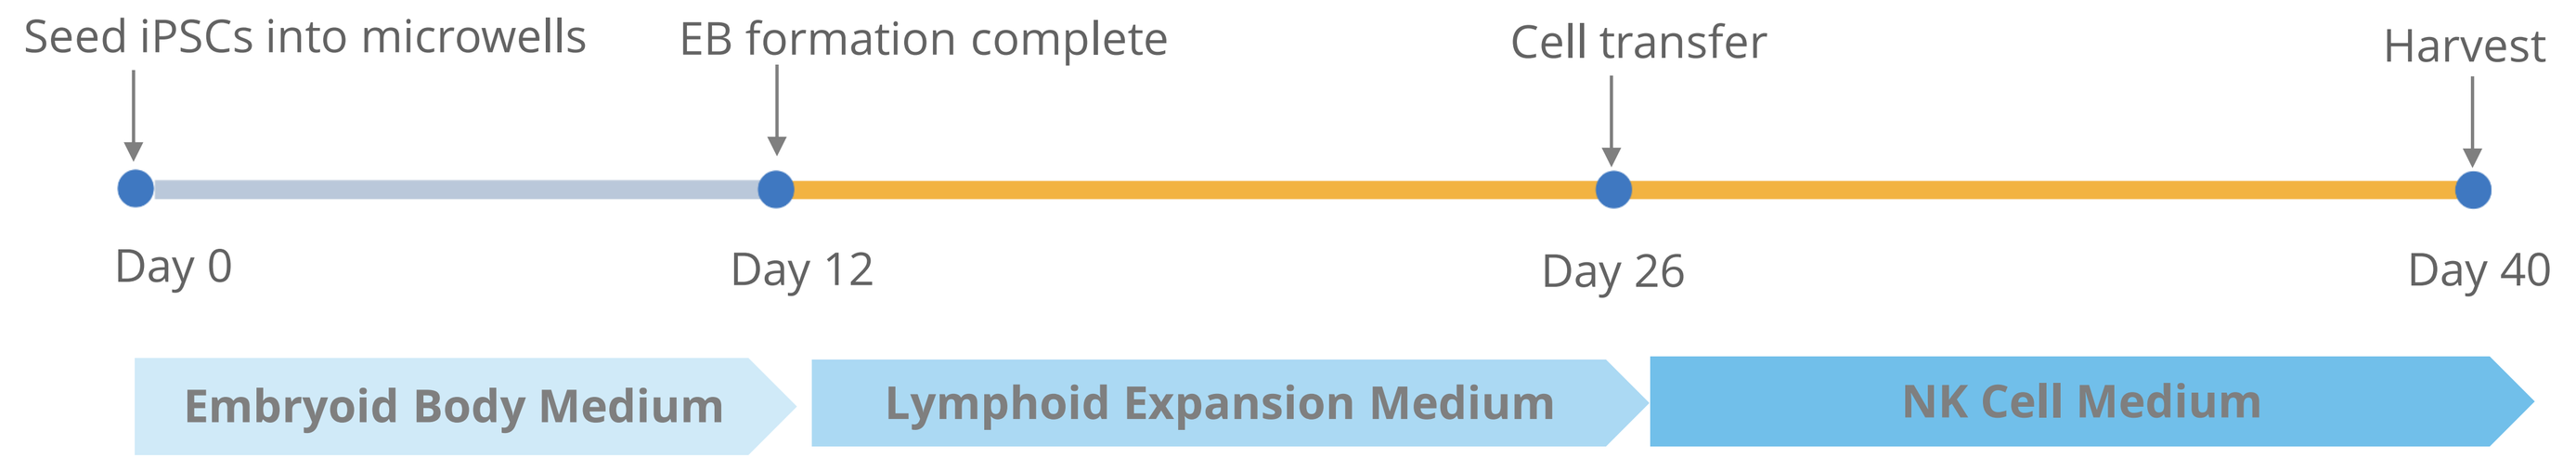
Seed iPSCs into microwells
EB formation complete
Cell transfer
Harvest
Day 0
Day 12
Day 26
Day 40
Embryoid Body Medium
Lymphoid Expansion Medium
NK Cell Medium

Figure 7. Differentiation of iPSCs into lymphocytes. iPSCs were differentiated into EBs following the steps outlined in Figure 4. On day 12, EBs were enzymatically dissociated into single cells and subsequently underwent magnetic bead selection to isolate CD34+ cells. Selected cells were seeded into Lymphoid Progenitor Expansion Medium (STEMCELL Technologies), followed by NK Cell Differentiation Medium (STEMCELL Technologies), and then characterized.

Isolation of CD34+ cells from iPSC-derived embryoid bodies

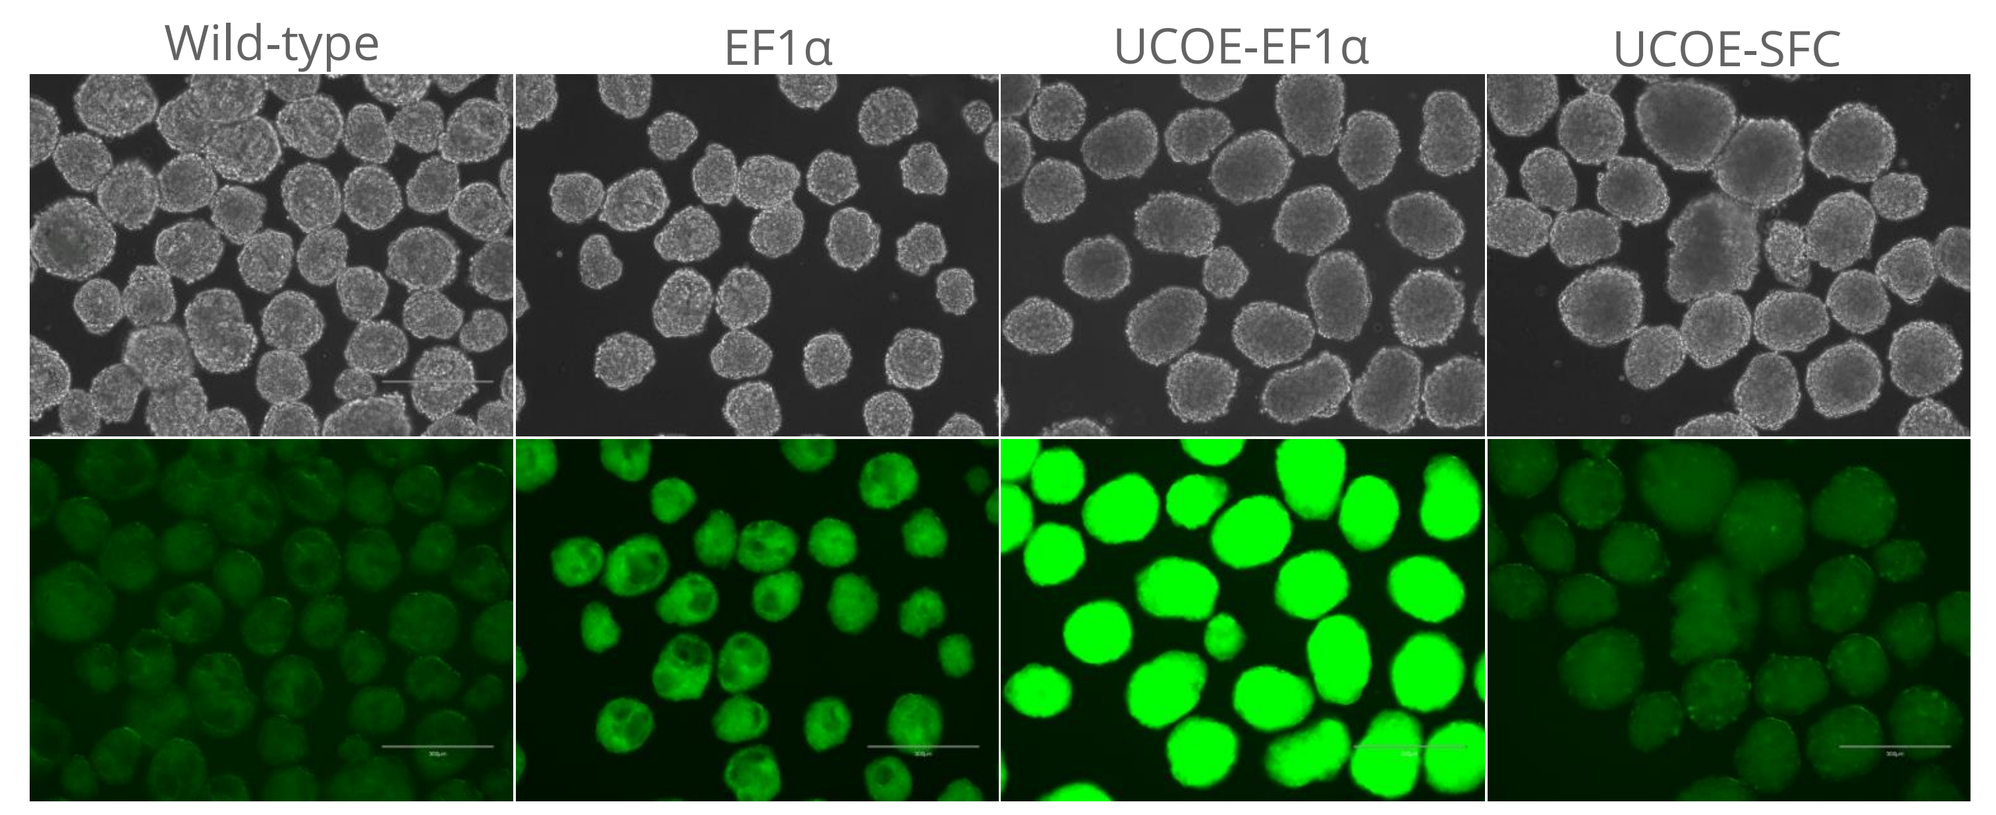
(8a)
Wild-type
EF1α
UCOE-EF1α
UCOE-SFC

(8b)

% of CD34+ cells on D12	
Wild-type	15.0%
EF1α	12.4%
UCOE-EF1α	9.4%
UCOE-SFC	11.9%

Figure 8. Dissociation of embryoid bodies and isolation of CD34+ cells. Figure 8a. Brightfield (top) and GFP (bottom) images of iPSC-derived embryoid bodies on day 5 of differentiation. Figure 8b. Percentage of CD34+ cells recovered after enzymatic dissociation of embryoid bodies on day 12, assessed using cell count after CD34 selection.

iLymphocytes with UCOEs display higher transgene expression

Figure 9. Transgene expression in iLymphocytes on day 40. Left: Percentage of cells expressing GFP, assessed using flow cytometry, on days 0 and 40 of differentiation. Error bars represent SEM. Right: Relative median fluorescence intensity (MFI) of GFP+ cells on day 40 of differentiation.

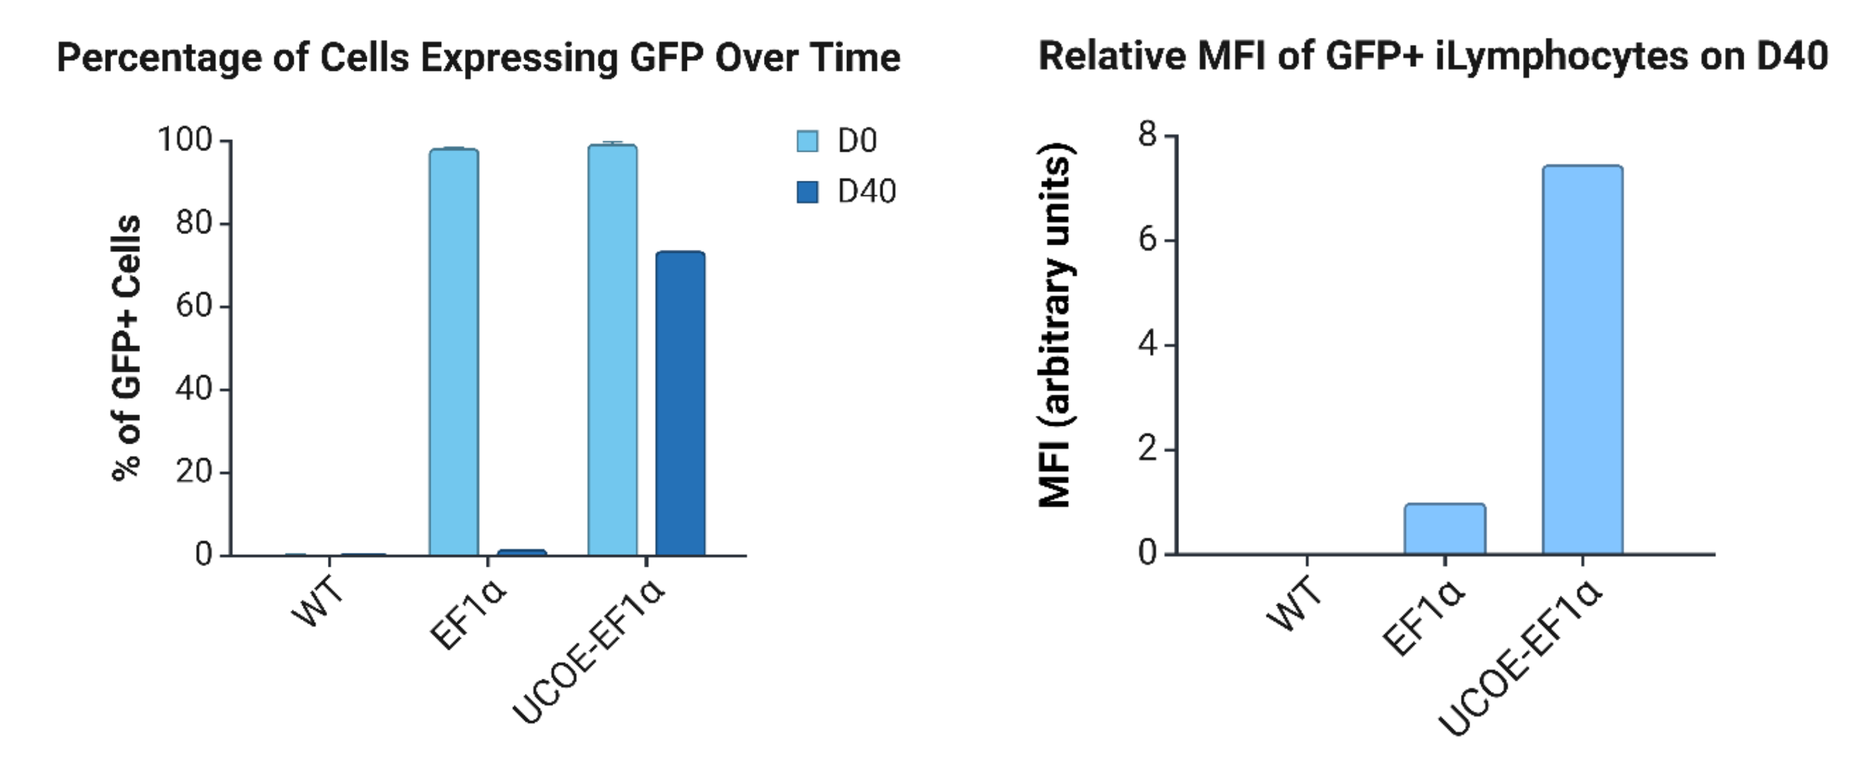
Percentage of Cells Expressing GFP Over Time
% of GFP+ Cells
D0
D40
WT
EF1α
UCOE-EF1α
Relative MFI of GFP+ iLymphocytes on D40
MFI (arbitrary units)
8
6
4
2
0

Assessment of lymphocyte and natural killer cell markers

Figure 10. Assessment of lymphocyte and natural killer (NK) cell markers on day 40 of differentiation. Expression of CD7 (early lymphocyte marker) and CD56 (classical NK cell marker) were evaluated using flow cytometry.

Surface marker	Wild-type	EF1α	UCOE-EF1α
CD7	77.7%	88.5%	54.5%
CD56	55.1%	28.9%	54.3%

American Society of Gene + Cell Therapy